The UCGI 28 PANIRINOX trial - a randomized phase II study assessing Panitumumab + FOLFIRINOX or mFOLFOX6 in RAS and BRAF wild type metastatic colorectal cancer patients (mCRC) selected from circulating DNA analysis

614TiP

Thibault Mazard MD-PhD(1), François Ghiringhelli MD-PhD(2), Caroline Mollevi PhD(1), Eric Assenat MD-PhD(3), Céline Gavoille MD(4), Denis Smith MD(5), Veronica Pezzella(6), Romain Meddeb PhD(7), Brice Pastor(7), Marc Ychou MD-PhD(1), Alain R. Thierry PhD(7)

[1]ICM-Val d'Aurelle, Montpellier-France [2]Centre Georges Francois Leclerc, Dijon– France [3]CHU Saint Eloi, Montpellier– France [4]Institut de Cancérologie de Lorraine, Vandoeuvre-les-Nancy- France [5]CHU de Bordeaux - Hôpital Haut Levèque, Pessac- France [6]R&D UNICANCER, Paris– France

Background and rationale

- A majority of patients with mCRC are not suitable for potentially curative resection and their management consists in palliative-intended chemotherapy (CT) [1].
- Nevertheless, it is described that in patients who achieve complete response (CR) with CT alone or after multimodality treatment, median overall survival is significantly longer than in other patients [2,3].
- In combination with cytotoxic doublet, anti-EGFR antibodies appear to be the biological agents of choice in order to reach the best response rate as long as tumors are RAS wild-type [4-6]. In this setting, FOLFOX plus panitumumab has become one of standard treatment since PRIME study results [7].
- In patients with good performance status, cytotoxic triplet combined to anti-EGFR therapies achieve impressive activity (>70% of objective response) and allow R0 secondary metastasectomies (rate from 28 to 35%) in several phase II studies [8-10]. Nevertheless, comparison of the association anti-EGFR with doublet versus triplet backbone is still missing.
- To determine RAS and BRAF mutational status, we have already demonstrated the clinical validity and utility of circulating DNA (ctDNA) analysis using IntPlex® method:
 - in a prospective multicenter study, gathering 106 mCRC patients before anti-EGFR therapy initiation, ctDNA was detected in 100% of patients. For the seven tested KRAS point mutations, the method exhibited 98% specificity and 92% sensitivity with a concordance value of 96% compared to tissue routine gold-standard methods. It also showed 100% specificity and sensitivity for the BRAF V600E mutation [11].
 - in a real-time blinded prospective multicenter clinical study, including 140 metastatic colorectal patients, we then concluded that ctDNA analysis advantageously reduced turnaround-time before result communication compared to tumour-tissue analysis (median time=2 days vs 12 days). When no threshold is applied, ctDNA analysis also revealed much more mutations thereby more accurately reflecting the real-time tumor biology [12].

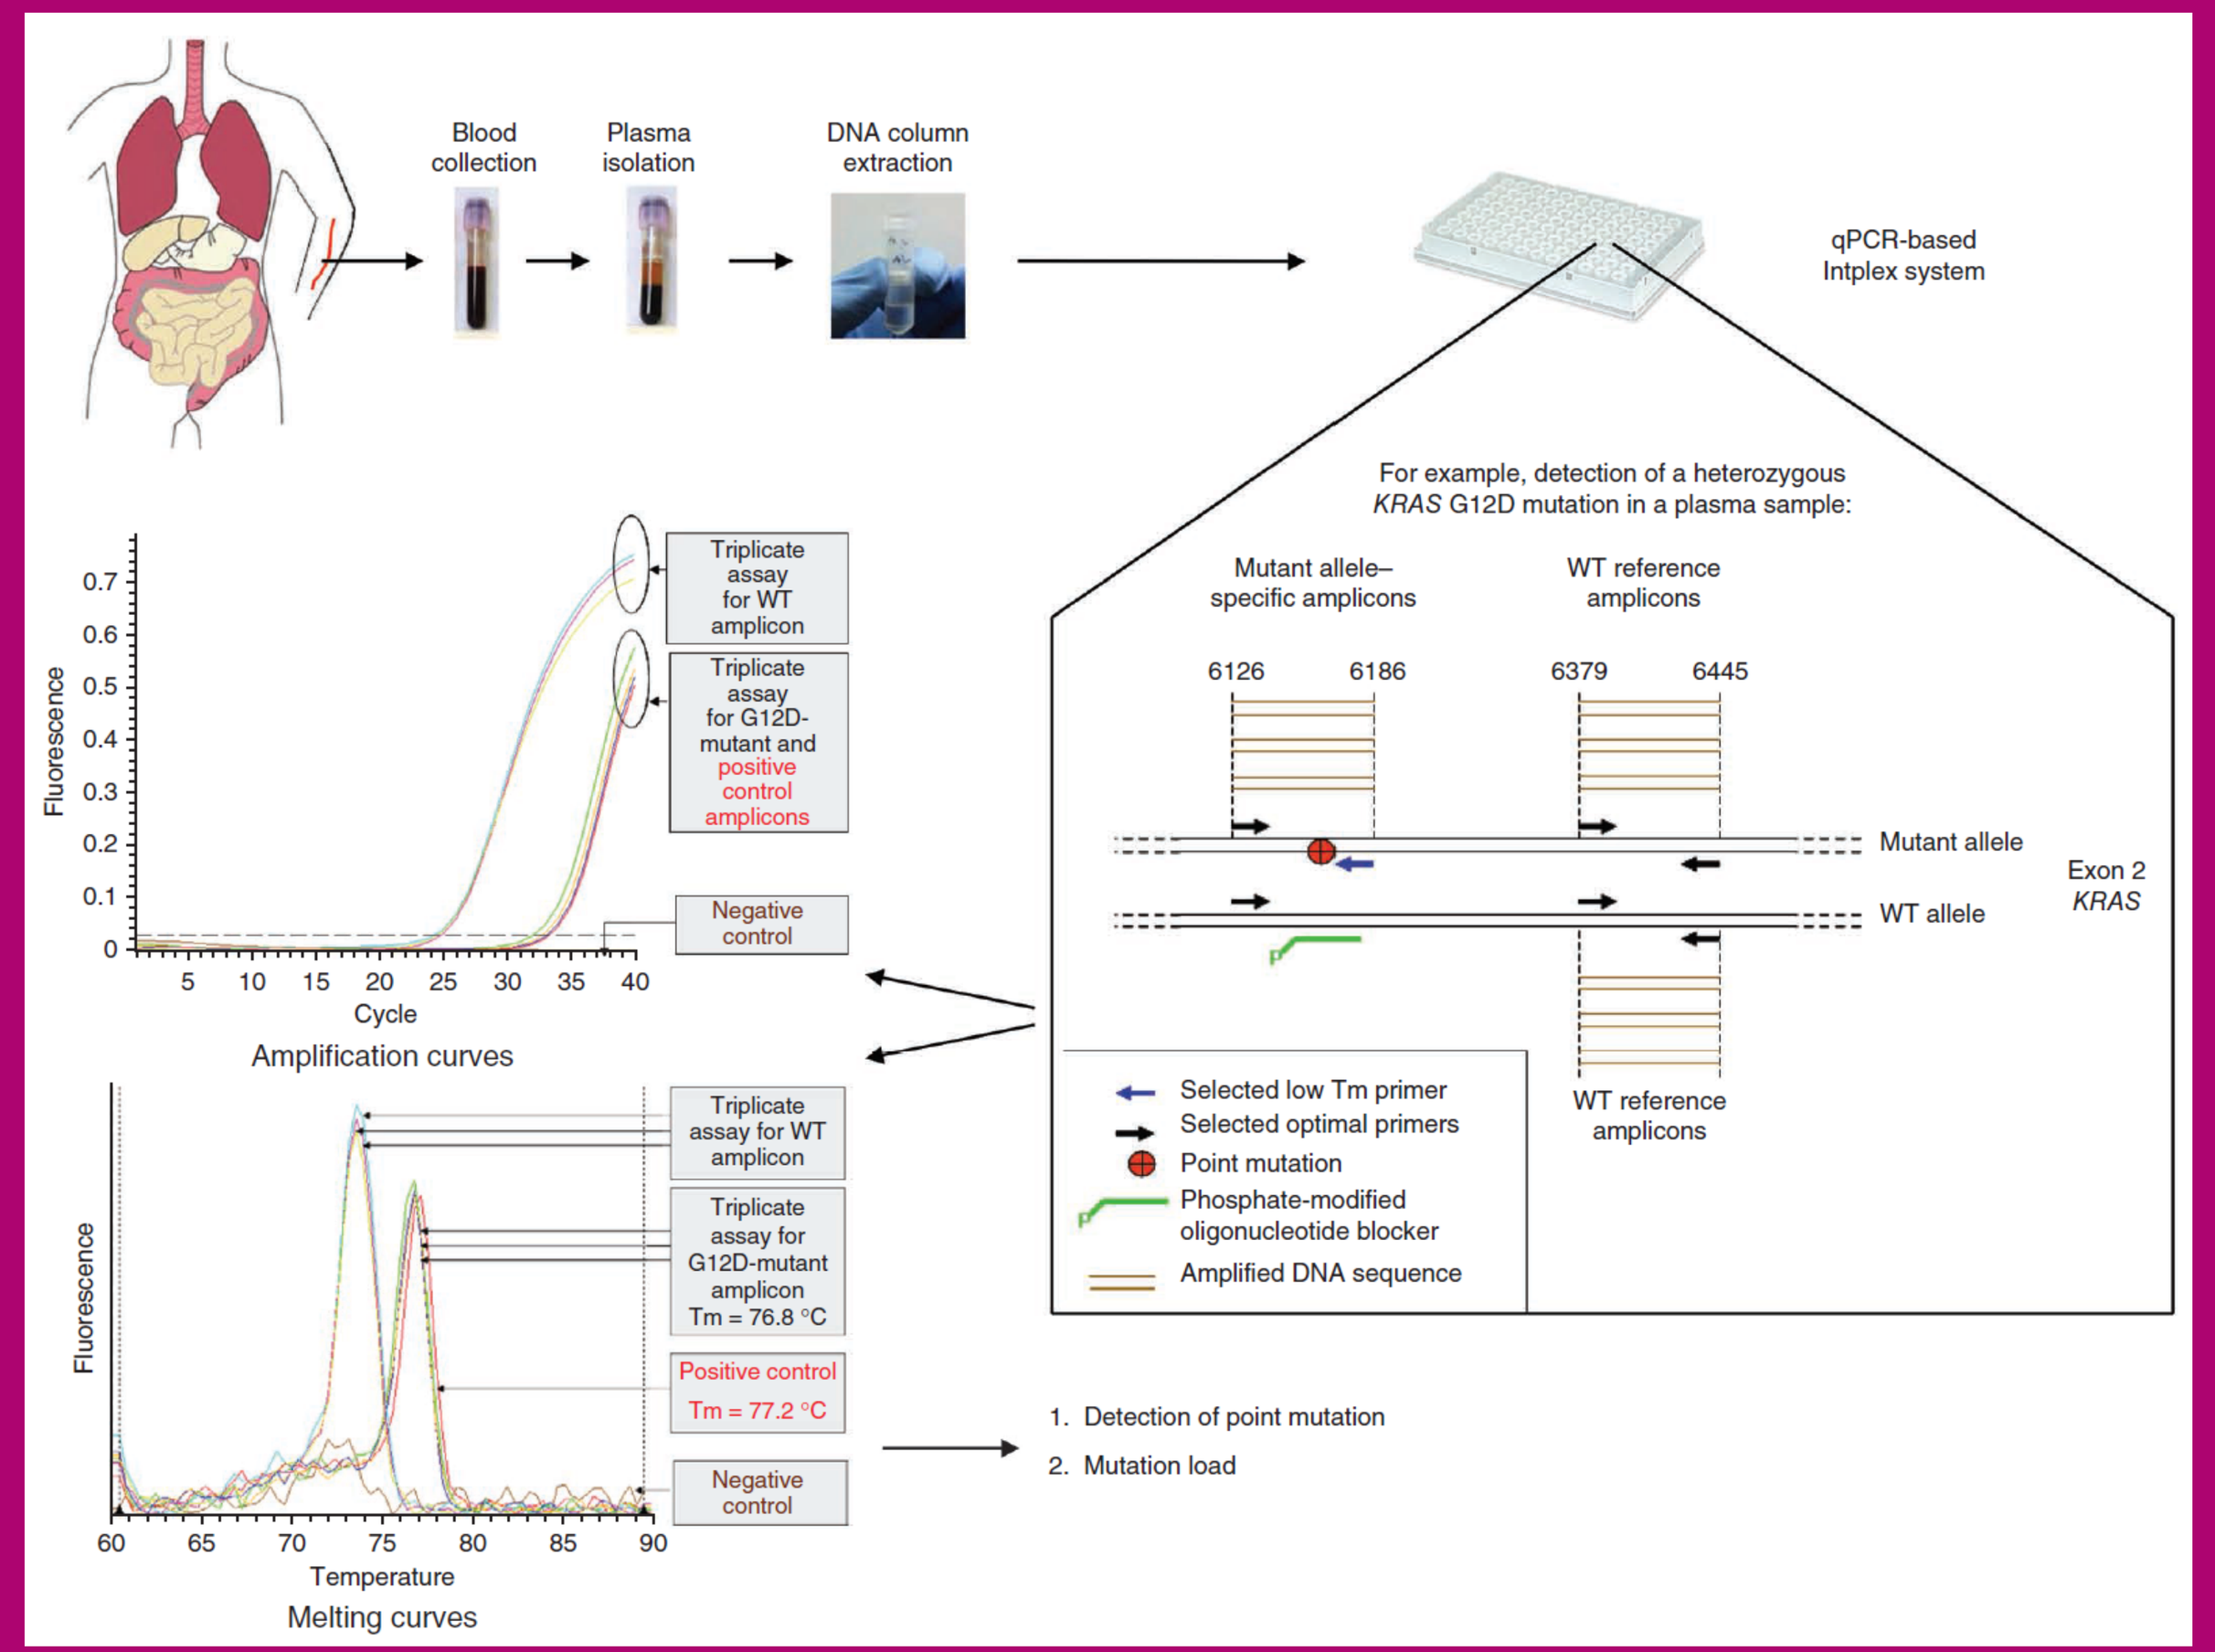
Blood collection
Plasma isolation
DNA column extraction
qPCR-based Intplex system
For example, detection of a heterozygous KRAS G12D mutation in a plasma sample:
Mutant allele–specific amplicons
WT reference amplicons
Mutant allele
WT allele
Exon 2 KRAS
Triplicate assay for WT amplicon
Triplicate assay for G12D-mutant and positive control amplicons
Negative control
Amplification curves
Cycle
Fluorescence
Selected low Tm primer
Selected optimal primers
Point mutation
Phosphate-modified oligonucleotide blocker
Amplified DNA sequence
Triplicate assay for G12D-mutant amplicon Tm = 76.8 °C
Positive control Tm = 77.2 °C
1. Detection of point mutation
2. Mutation load
Temperature
Melting curves

Schematics of the methodology and the analysis of point mutation detection from plasma by the Intplex system based on the Allele-Specific qPCR method Applied for KRAS G12D mutation, from Thierry et al, Nature Med 2014

- Selecting patients with this technology, we aim to investigate response rate and outcomes reached with panitumumab in combination with a standard (mFOLFOX6) or an intensified CT regimen (FOLFIRINOX) in RAS and BRAF wild-type (WT) metastatic patients.

Study design

- **PANIRINOX** (**ClinicalTrials.gov ID: NCT02980510**) is a national multicentric open-label randomized phase II trial assessing FOLFIRINOX + Panitumumab versus mFOLFOX6 + Panitumumab in metastatic colorectal cancer patients selected by RAS and BRAF status from ctDNA analysis.
- Patients will be randomized 2:1 to either FOLFIRINOX-panitumumab or mFOLFOX6-panitumumab with 12 planned cycles in each arm.
- For each arm, patients are stratified according to disease-extent (non/liver limited disease).
- Its design is summarized below:

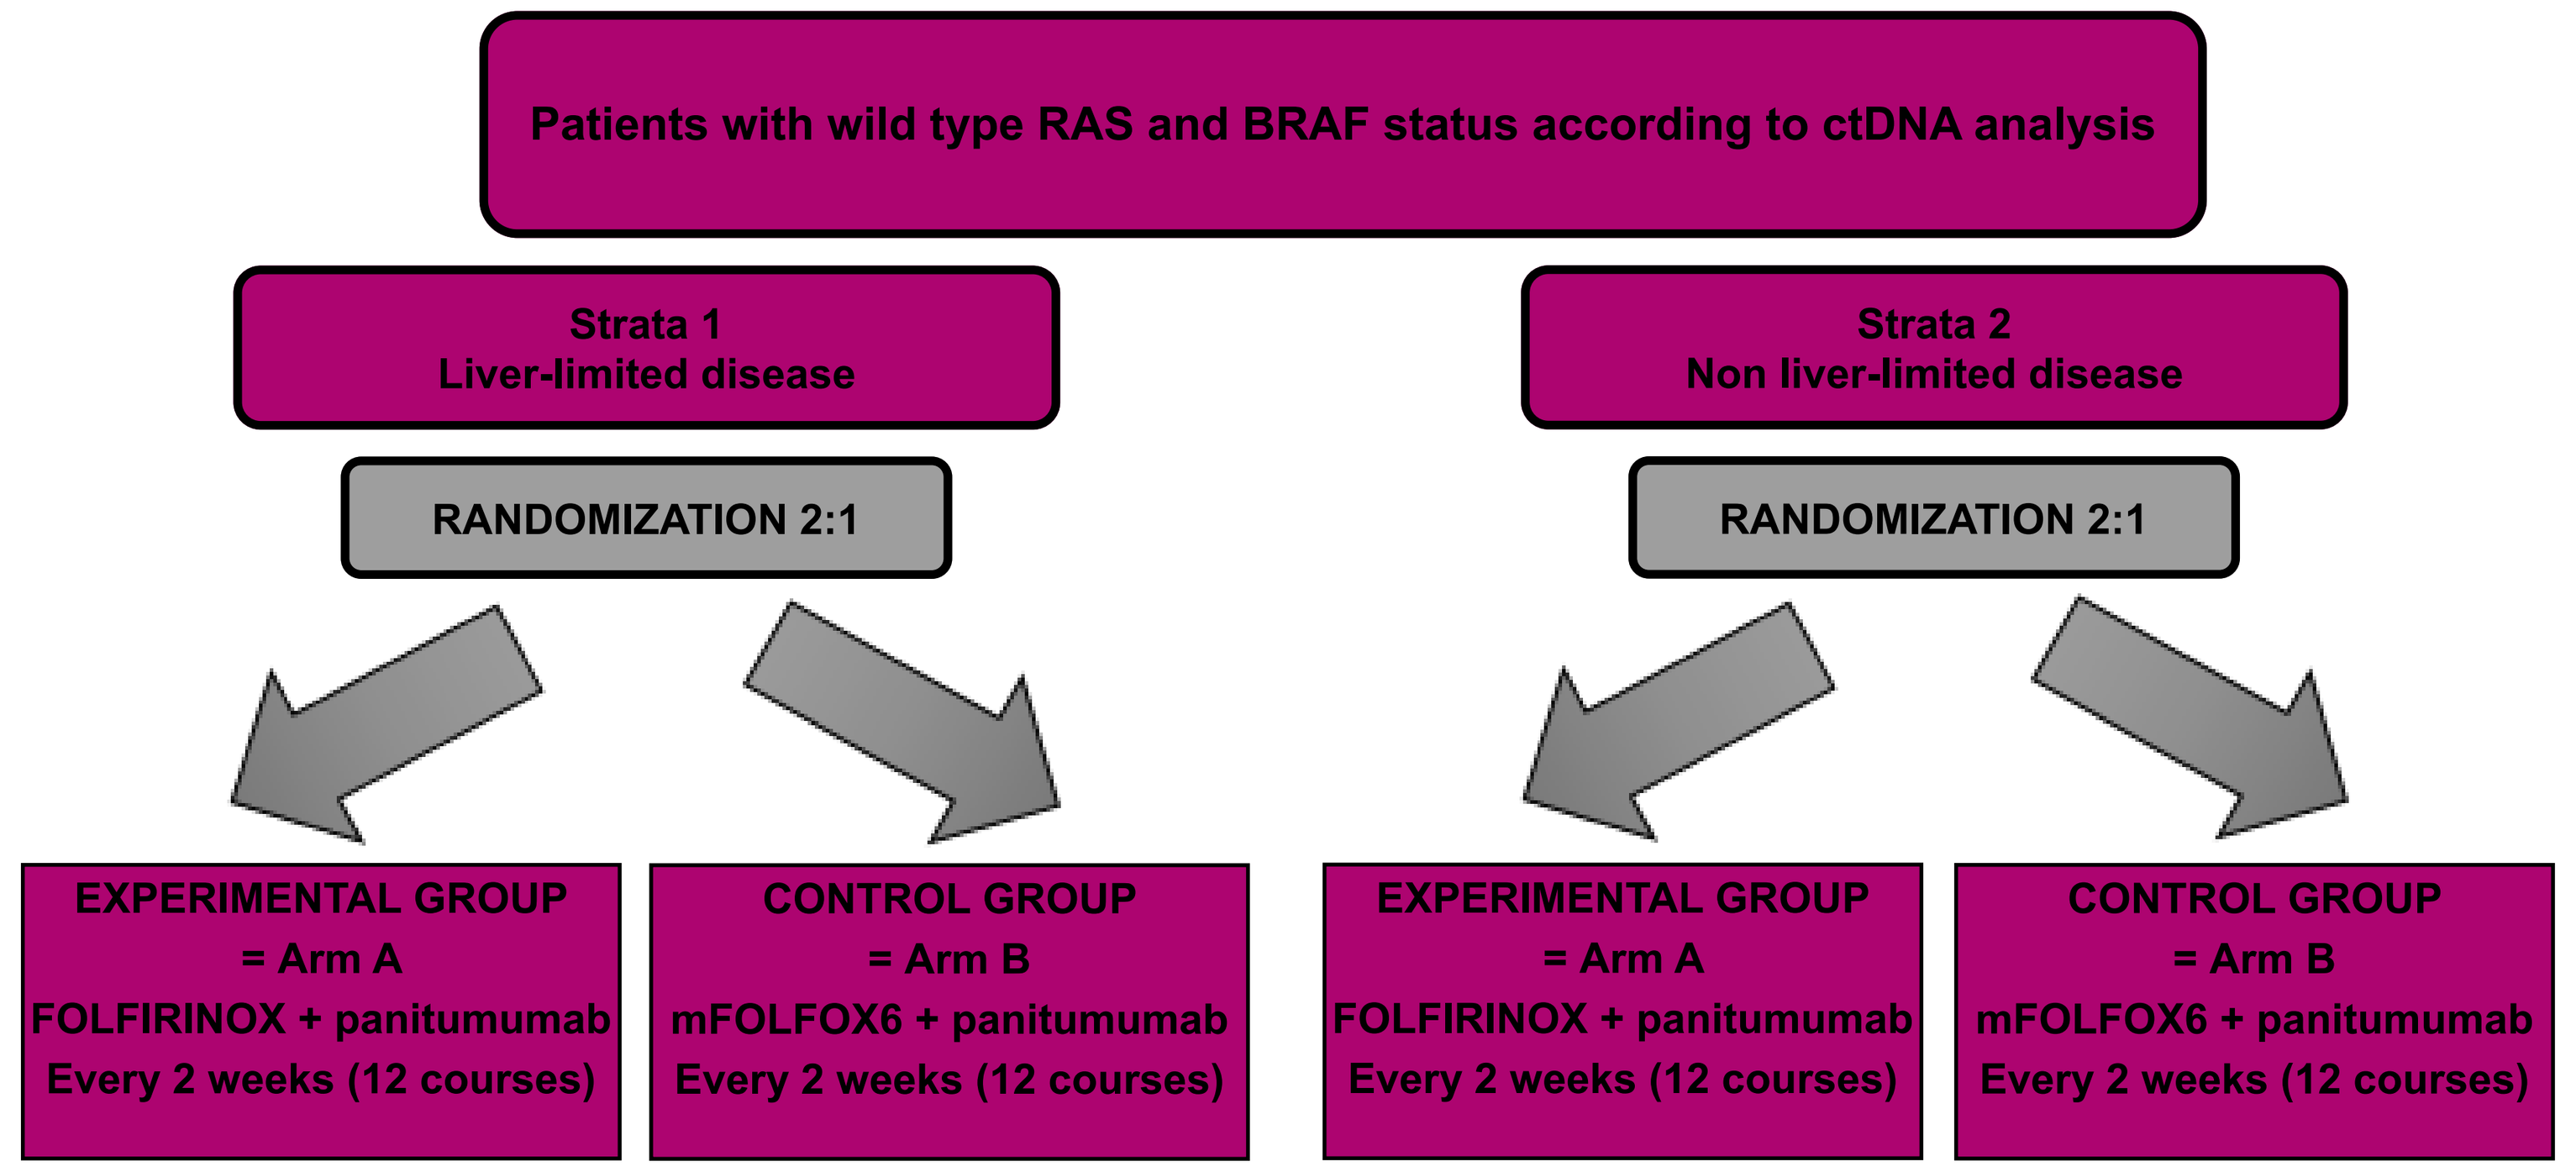
Patients with wild type RAS and BRAF status according to ctDNA analysis
Strata 1
Liver-limited disease
Strata 2
Non liver-limited disease
RANDOMIZATION 2:1
RANDOMIZATION 2:1
EXPERIMENTAL GROUP = Arm A FOLFIRINOX + panitumumab Every 2 weeks (12 courses)
CONTROL GROUP = Arm B mFOLFOX6 + panitumumab Every 2 weeks (12 courses)
EXPERIMENTAL GROUP = Arm A FOLFIRINOX + panitumumab Every 2 weeks (12 courses)
CONTROL GROUP = Arm B mFOLFOX6 + panitumumab Every 2 weeks (12 courses)

In each arm, **Panitumumab** over 1 hour at 6mg/kg every 2 weeks on day 1 before:

-**FOLFIRINOX** : oxaliplatin 85mg/m² IV infusion over 2 hours immediately followed by folinic acid 400mg/m² given as a 2-hour IV infusion with the addition, after 30 minutes of irinotecan 150mg/m² given as a 90-minute intravenous infusion through a Y-connector immediately followed by Fluorouracil 400mg/m² IV bolus then 5-FU 2400 mg/m² over 46 hours continuous infusion.

-or, **mFOLFOX6:** oxaliplatin 85mg/m² IV infusion over 2 hours immediately followed by folinic acid 400mg/m² IV infusion over 2 hours followed by fluorouracil 400mg/m² IV bolus then 5-FU 2400mg/m² over 46 hours continuous infusion.

Main Inclusion Criteria

- Age between 18 and 75 years
- ECOG PS between 0 and 1
- Histologically confirmed colo-rectal adenocarcinoma
- Untreated synchronous or metachronous metastatic disease deemed unresectable with curative intent
- KRAS(codons 12, 13, 59, 61, 117, 146), NRAS(codons 12, 13, 59, 61) and BRAF (codon 600) WT tumor status according to plasma analysis of ctDNA by Intplex technology
- Measurable disease according to RECIST version 1.1
- Adequate hematologic, hepatic and renal functions

Main Exclusion Criteria

- Adjuvant treatment with oxaliplatin
- Previous treatment for metastatic disease
- Brain metastases
- Patients with a history of severe or life-threatening hypersensitivity to the active substances or to any of the excipients delivered in this study
- Previous organ transplantation, HIV or other immunodeficiency syndromes
- Concomitant medications/comorbidities that may prevent the patient from receiving study treatment
- History of other malignancy within the previous 5 years
- Other concomitant cancer

Objectives

Primary objective

To evaluate in both arms:

- Complete response rate

Secondary objectives

To assess in both arms:

- Overall Survival (OS)
- Progression Free Survival (PFS)
- Secondary resection
- Early tumor shrinkage (ETS)
- Depth of response (DpR)
- Safety profile
- Diagnostic performance of ctDNA analysis compared to the tumor-tissue analysis (current gold standard)

Ancillary objective:

To detect in both arms:

- Emergence of ctDNA RAS and BRAF mutations under anti-EGFR-based chemotherapy

Primary endpoint

Complete response=complete disappearance of metastatic lesions according to RECIST 1.1 and CEA level normalization (if initially increased) after a maximum of 12 cycles of chemotherapy.
It can be reached with chemotherapy only or with a multimodal approach (surgical resection, regional procedures as radiofrequency, cryoablation, radiation therapy).

Required number of patients

- **Strata 1 (liver-limited disease):**

One-stage Fleming design with α=5%, β=10%, p0 (the probability of inefficiency maximum)=20%, p1 (the probability of minimum efficiency)=35%
=>72 evaluable patients (76 patients/ 5% non-evaluable patients).
FOLFIRINOX-panitumumab sufficiently effective if at least 20 successes (complete response) out of 72 evaluable patients.

mFOLFOX6+panitumumab=internal control to validate the hypothesis of a complete response rate equal to 20% in the strata 1 (liver limited disease). Calculation of 95% Confidence Interval (95%CI) on 38 patients of the control group.

- **Strata 2 (non-liver-limited disease)**:

One-stage Fleming design with α=5%, β=10%, p0 (the probability of inefficiency maximum)=3%, p1 (the probability of minimum efficiency)=12%
=>60 evaluable patients (63 patients/ 5% non-evaluable patients).
FOLFIRINOX-panitumumab sufficiently effective if at least 4 successes (complete response) out of 60 evaluable patients.

mFOLFOX6+panitumumab=internal control to validate the hypothesis of a complete response rate equal to 3% in the strata 2 (non-liver-limited disease). Calculation of 95% Confidence Interval (95%CI) on 32 patients of the control group.

Total of 209 patients included: 139 patients in the FOLFIRINOX+panitumumab group (Strata 1 N=76 and Strata 2 N=63) and 70 patients in the mFOLFOX6+panitumumab group (Strata 1 N=38 and Strata 2 N=32).

Assessments

- Before inclusion, RAS and BRAF status are determined, by Dr Alain Thierry's team, according to plasma analysis of ctDNA by Intplex® technology. Applying the threshold used in the first study that validated the test[11], patient is considered as mutated if ctDNA harbors a mutational load (mutation allelic frequency=mA%) higher than 0.5%.
- Response to therapy is evaluated by measuring changes in tumor size by CT Scan and/or MRI, according to the RECIST v1.1 criteria, every 4 cycles.
- Every complete response has to be confirmed 4 to 6 weeks after the last treatment and by no residual disease activity on PET Scan done 3 months after the end of the last treatment.
- A central review of CT-Scans and/or MRIs, PET Scans will be performed in patients who reach a complete response.
- Adverse events are graded according to the National CTCAE version 4.03.
- For ancillary study, blood samples will be collected every 4 cycles and analyzed with Intplex® technology in order to detect the occurrence of RAS and BRAF hotspot mutations in ctDNA.

Current status

- Number of opened sites= 10
- Number of expected enrollement sites= 20-30
- Number of screened patients= 54
- Number of recruited patients= 14
- Number of expected patients= 209 randomized/464 screened
- Inclusion period= 3 years

Expected results

- This study will be the **first interventional study using cfDNA analysis,** in the whole oncology field, for selecting patients. It could definitely validate its use in daily practice.
- It will be the **first study to directly compare activity of triplet vs doublet chemotherapy backbone in combination with panitumumab**, as upfront treatment, in highly molecularly selected unresectable mCRC patients.

References

1- Van Cutsem E et al. Annals of Oncology 2016;27:1386–422. doi:10.1093/annonc/mdw235
2- Dy GK et al. Journal of Clinical Oncology 2007;25:3469–74. doi:10.1200/JCO.2007.10.7128
3- Ferrarotto R et al. Clinical Colorectal Cancer 2011;10:178–82. doi:10.1016/j.clcc.2011.03.023
4- Schwartzberg et al. Colorectal Cancer. Journal of Clinical Oncology 2014;32:2240–7. doi:10.1200/JCO.2013.53.2473
5- Heinemann V et al. The Lancet Oncology 2014;15:1065–1075.
6- Venook AP et al. JAMA 2017;317:2392. doi:10.1001/jama.2017.7105
7- Douillard JY et al. Annals of Oncology 2014;25:1346–55. doi:10.1093/annonc/mdu141
8- Assenat E et al. The Oncologist 2011;16:1557–64. doi:10.1634/theoncologist.2011-0141
9- Fornaro L et al. Annals of Oncology 2013;24:2062–7. doi:10.1093/annonc/mdt165
10- Cremolini C et al. JAMA Oncology 2018;4:529. doi:10.1001/jamaoncol.2017.5314
11- Thierry AR et al. Nature Medicine 2014;20(4):430-5. doi: 10.1038/nm.3511
12- Thierry AR et al. Annals of Oncology 2017;28:2149–59. doi:10.1093/annonc/mdx330

Acknowledgment

Patients and families
All the participating centers and investigational teams in France
Data management team at Institut de Cancérologie de Montpellier
AMGEN

Corresponding author's e-mail: Thibault.Mazard@icm.unicancer.fr