ESMO 2018, Munich
781 TiP

PRODIGE 29 - UCGI 26 - NEOPAN: - A Randomised trial of chemotherapy with Folfirinox or gemcitabine in locally advanced pancreatic carcinoma

M.P. Ducreux[1], R. Desgrippes[2], M. Texier[3], F. Di Fiore[4], P. Vanelslander[5], O. Capitain[6], R. Guimbaud[7], J. Forestier[8], F. El Hajbi[9], P. Follana[10], A. Parzy[11], N. Baba Hamed[12], Y. Rinaldi[13], T. Brotelle[14], J.-P. Pignon[3], C. Jouffroy[15], L. Dahan[16], O. Bouche[17]

[1]Gastroenterology, Gustave Roussy Cancer Campus, Villejuif, FR, [2]Medical oncology, C.H. Saint Malo, St. Malo, FR, [3]Biostatistics and epidemiology, Gustave Roussy Cancer Campus, Villejuif, FR, [4]Gastroenterology, CHU Hôpitaux de Rouen-Charles Nicolle, Rouen, FR, [5]Gastroenterology, Hospital Saint Claude, Saint Quentin, FR, [6]Medical oncology, Institut de Cancerologie de l'Ouest - Centre Paul Papin, Angers, FR [7]Digestive oncology, Centre Hospitalier Rangueil, Toulouse, FR, [8]Medical oncology, Hôpital Edouard Herriot, Lyon, FR, [9]Urology and digestive oncology, Centre Oscar Lambret, Lille, FR, [10]Medical oncology, Centre Anticancer Antoine Lacassagne, Nice, FR, [11]Digestive pathology, Centre Francois Baclesse, Caen, FR, [12]Oncology, Hopital St. Joseph, Paris, FR, [13]Oncology, Hôpital Européen, Marseille, FR, [14]Onco-hematology, Centre Hospitalier Henri Duffaut, Avignon, FR, [15]R&D, UNICANCER, Paris, FR, [16]Hepatogastroenterology and oncology department, CHU La Timone, Marseille, FR, [17]Hepatogastroenterology and oncology department, CHU de Reims - Hôpital Robert Debré, Reims, FR

Background

Pancreatic cancer (PC) is an aggressive malignancy and the 4th cause of all cancer deaths worldwide. More than 30% of patients with PC are unresectable because of the local extension with a median overall survival (OS) of less than one year. The standard of care remains gemcitabine alone for unresectable locally advanced PC.

It was demonstrated that FOLFIRINOX (5-FU, irinotecan, oxaliplatin) is superior to gemcitabine in the treatment of metastatic PC, in terms of OS and progression-free survival (PFS). The recent PRODIGE 24 study's results showed that mFOLFIRINOX is superior to gemcitabine in adjuvant therapy of resected PC, in terms of disease-free survival and OS. The superiority of FOLFIRINOX vs gemcitabine remains to be proven in unresectable locally advanced PC.

The aim of NEOPAN study is to evaluate the efficacy of FOLFIRINOX versus gemcitabine in the treatment of unresectable locally advanced PC.

Objectives

Primary objective :

- **To compare progression-free survival (PFS)** in patients with locally advanced adenocarcinoma of pancreas treated by Folfirinox or Gemcitabine

Secondary objectives :

To compare the following endpoints between the 2 treatment arms:

- **Composite index of induction treatment severe toxicity** (biliary tract infection Grade 3-4 + any grade 5 toxicities + induction chemotherapy interruption for toxicity) after 8 weeks of treatment
- Serious adverse events (NCI-CTC v4.0), compliance to treatment
- **Overall survival**
- Type of events from PFS
- Rate of surgery with curative intent (patients with R0 tumour resection)
- **Objective response rate and disease control rate** (RECIST 1.1 criteria)
- Time to treatment failure
- Quality of life (QLQ C30)

Participating sites

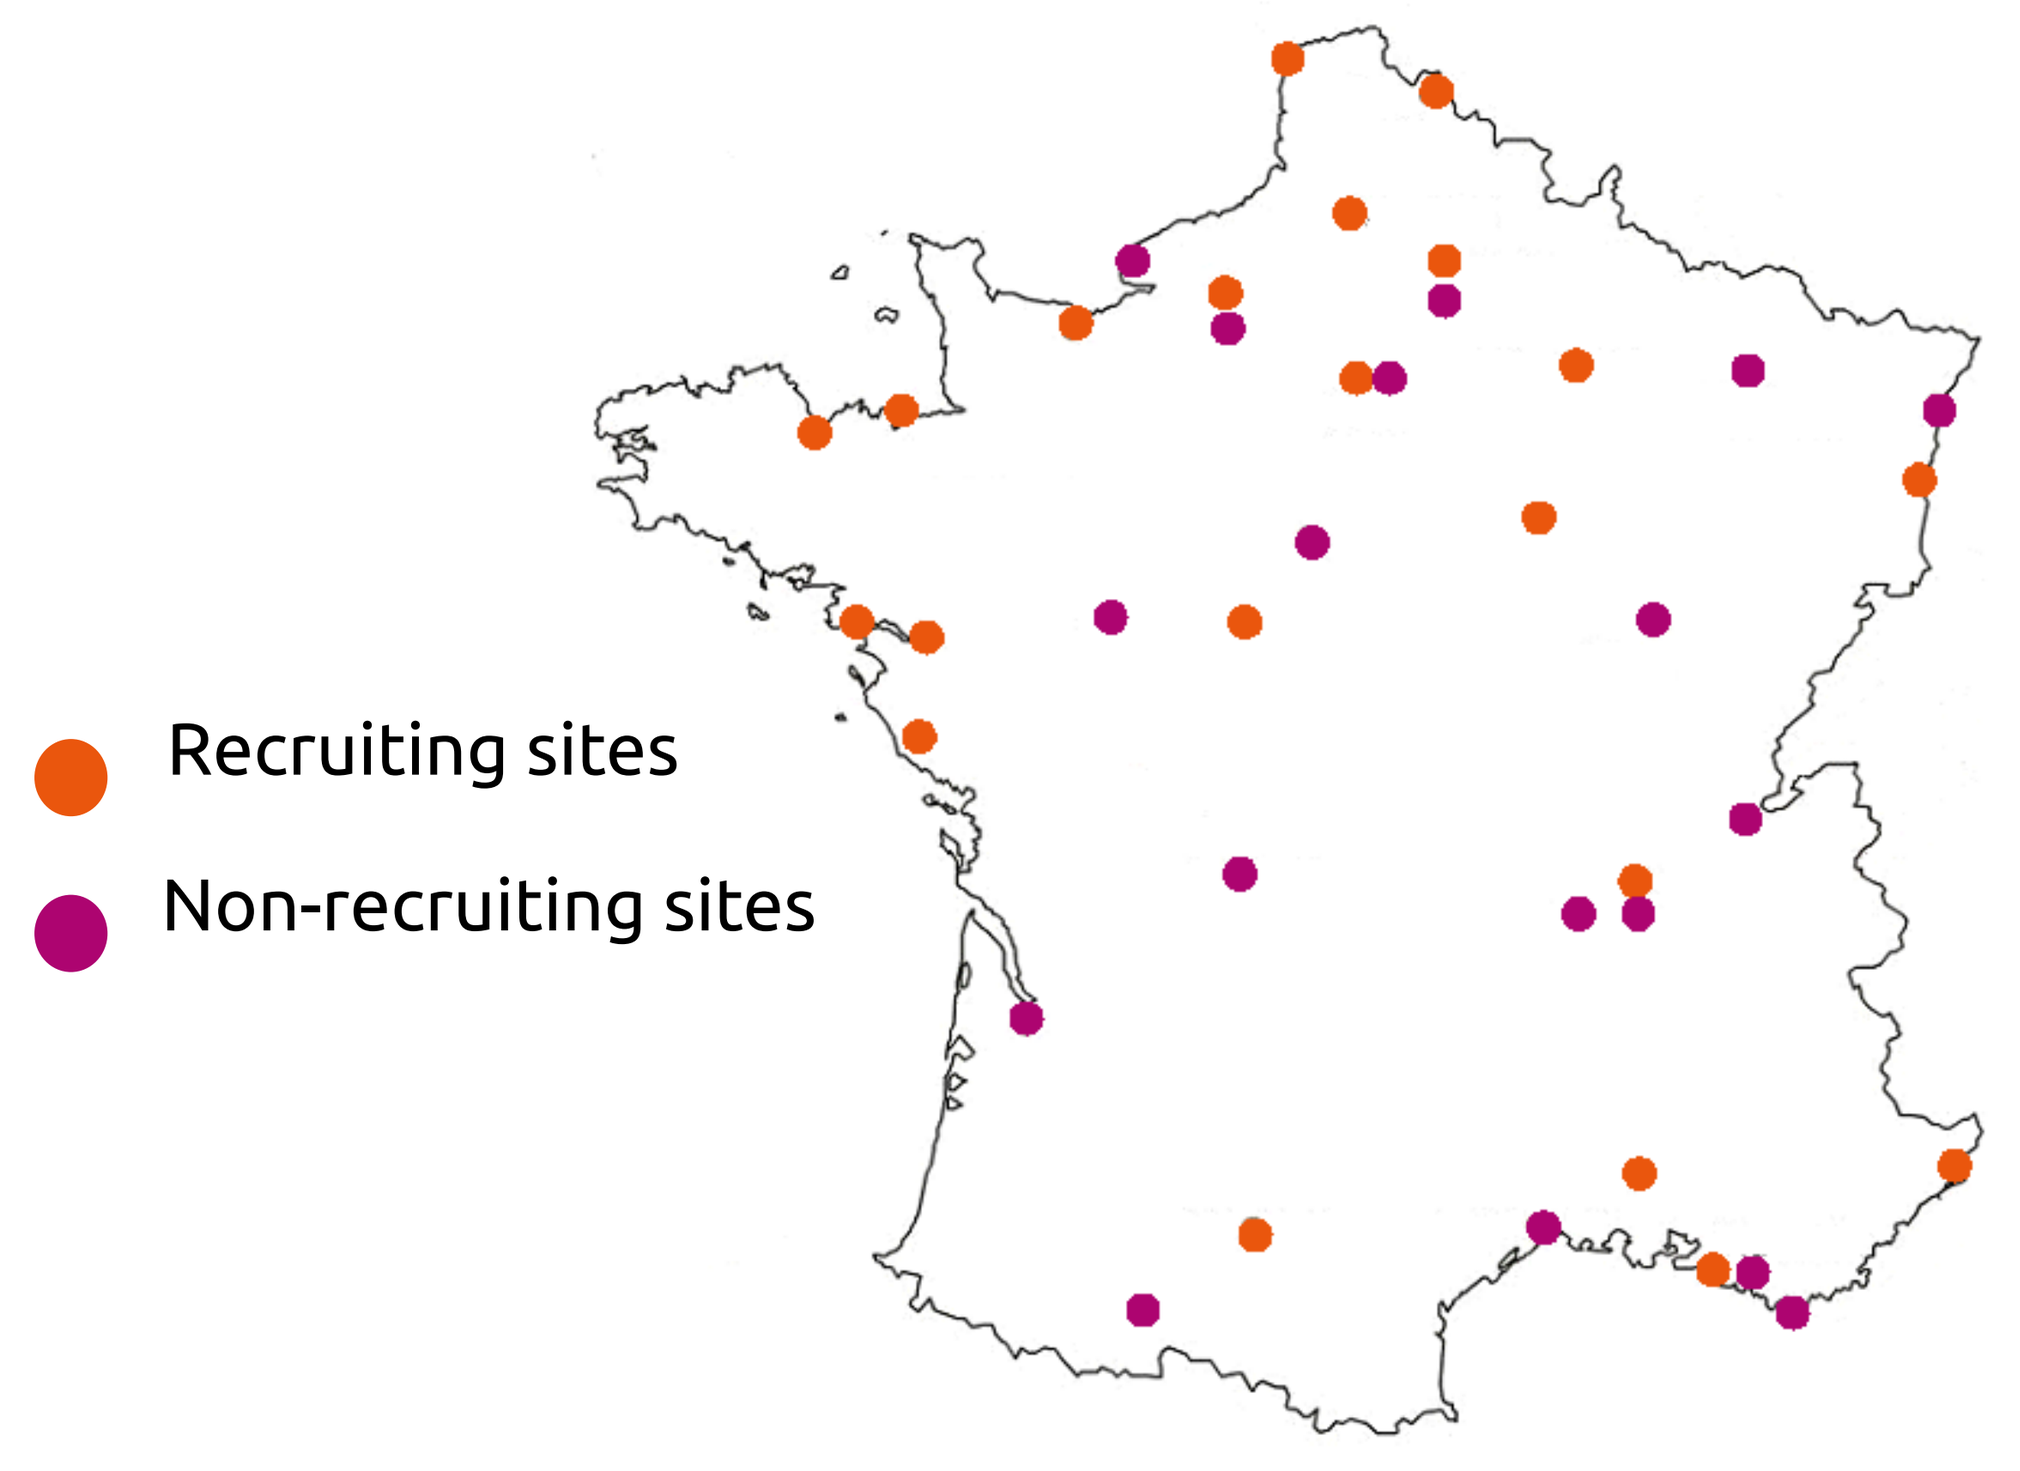
Recruiting sites
Non-recruiting sites

Figure 2 - Repartition of participating sites

Study Status

Study timelines

- Start of recruitment in Mar 2015
- Planned end of recruitment in March 2020
- Primary endpoint analysis expected in **March 2021**

IDMC meeting

- First IDMC in Jan 2018: Folfirinox toxicity was manageable and acceptable. Recommendation to continue the study without any change
- Next IDMC planned in Q1 2019

Accrual status

As of September 28th, 2018, **97** patients were identified over 27 centres. Among them **85** were randomized (figure 3).

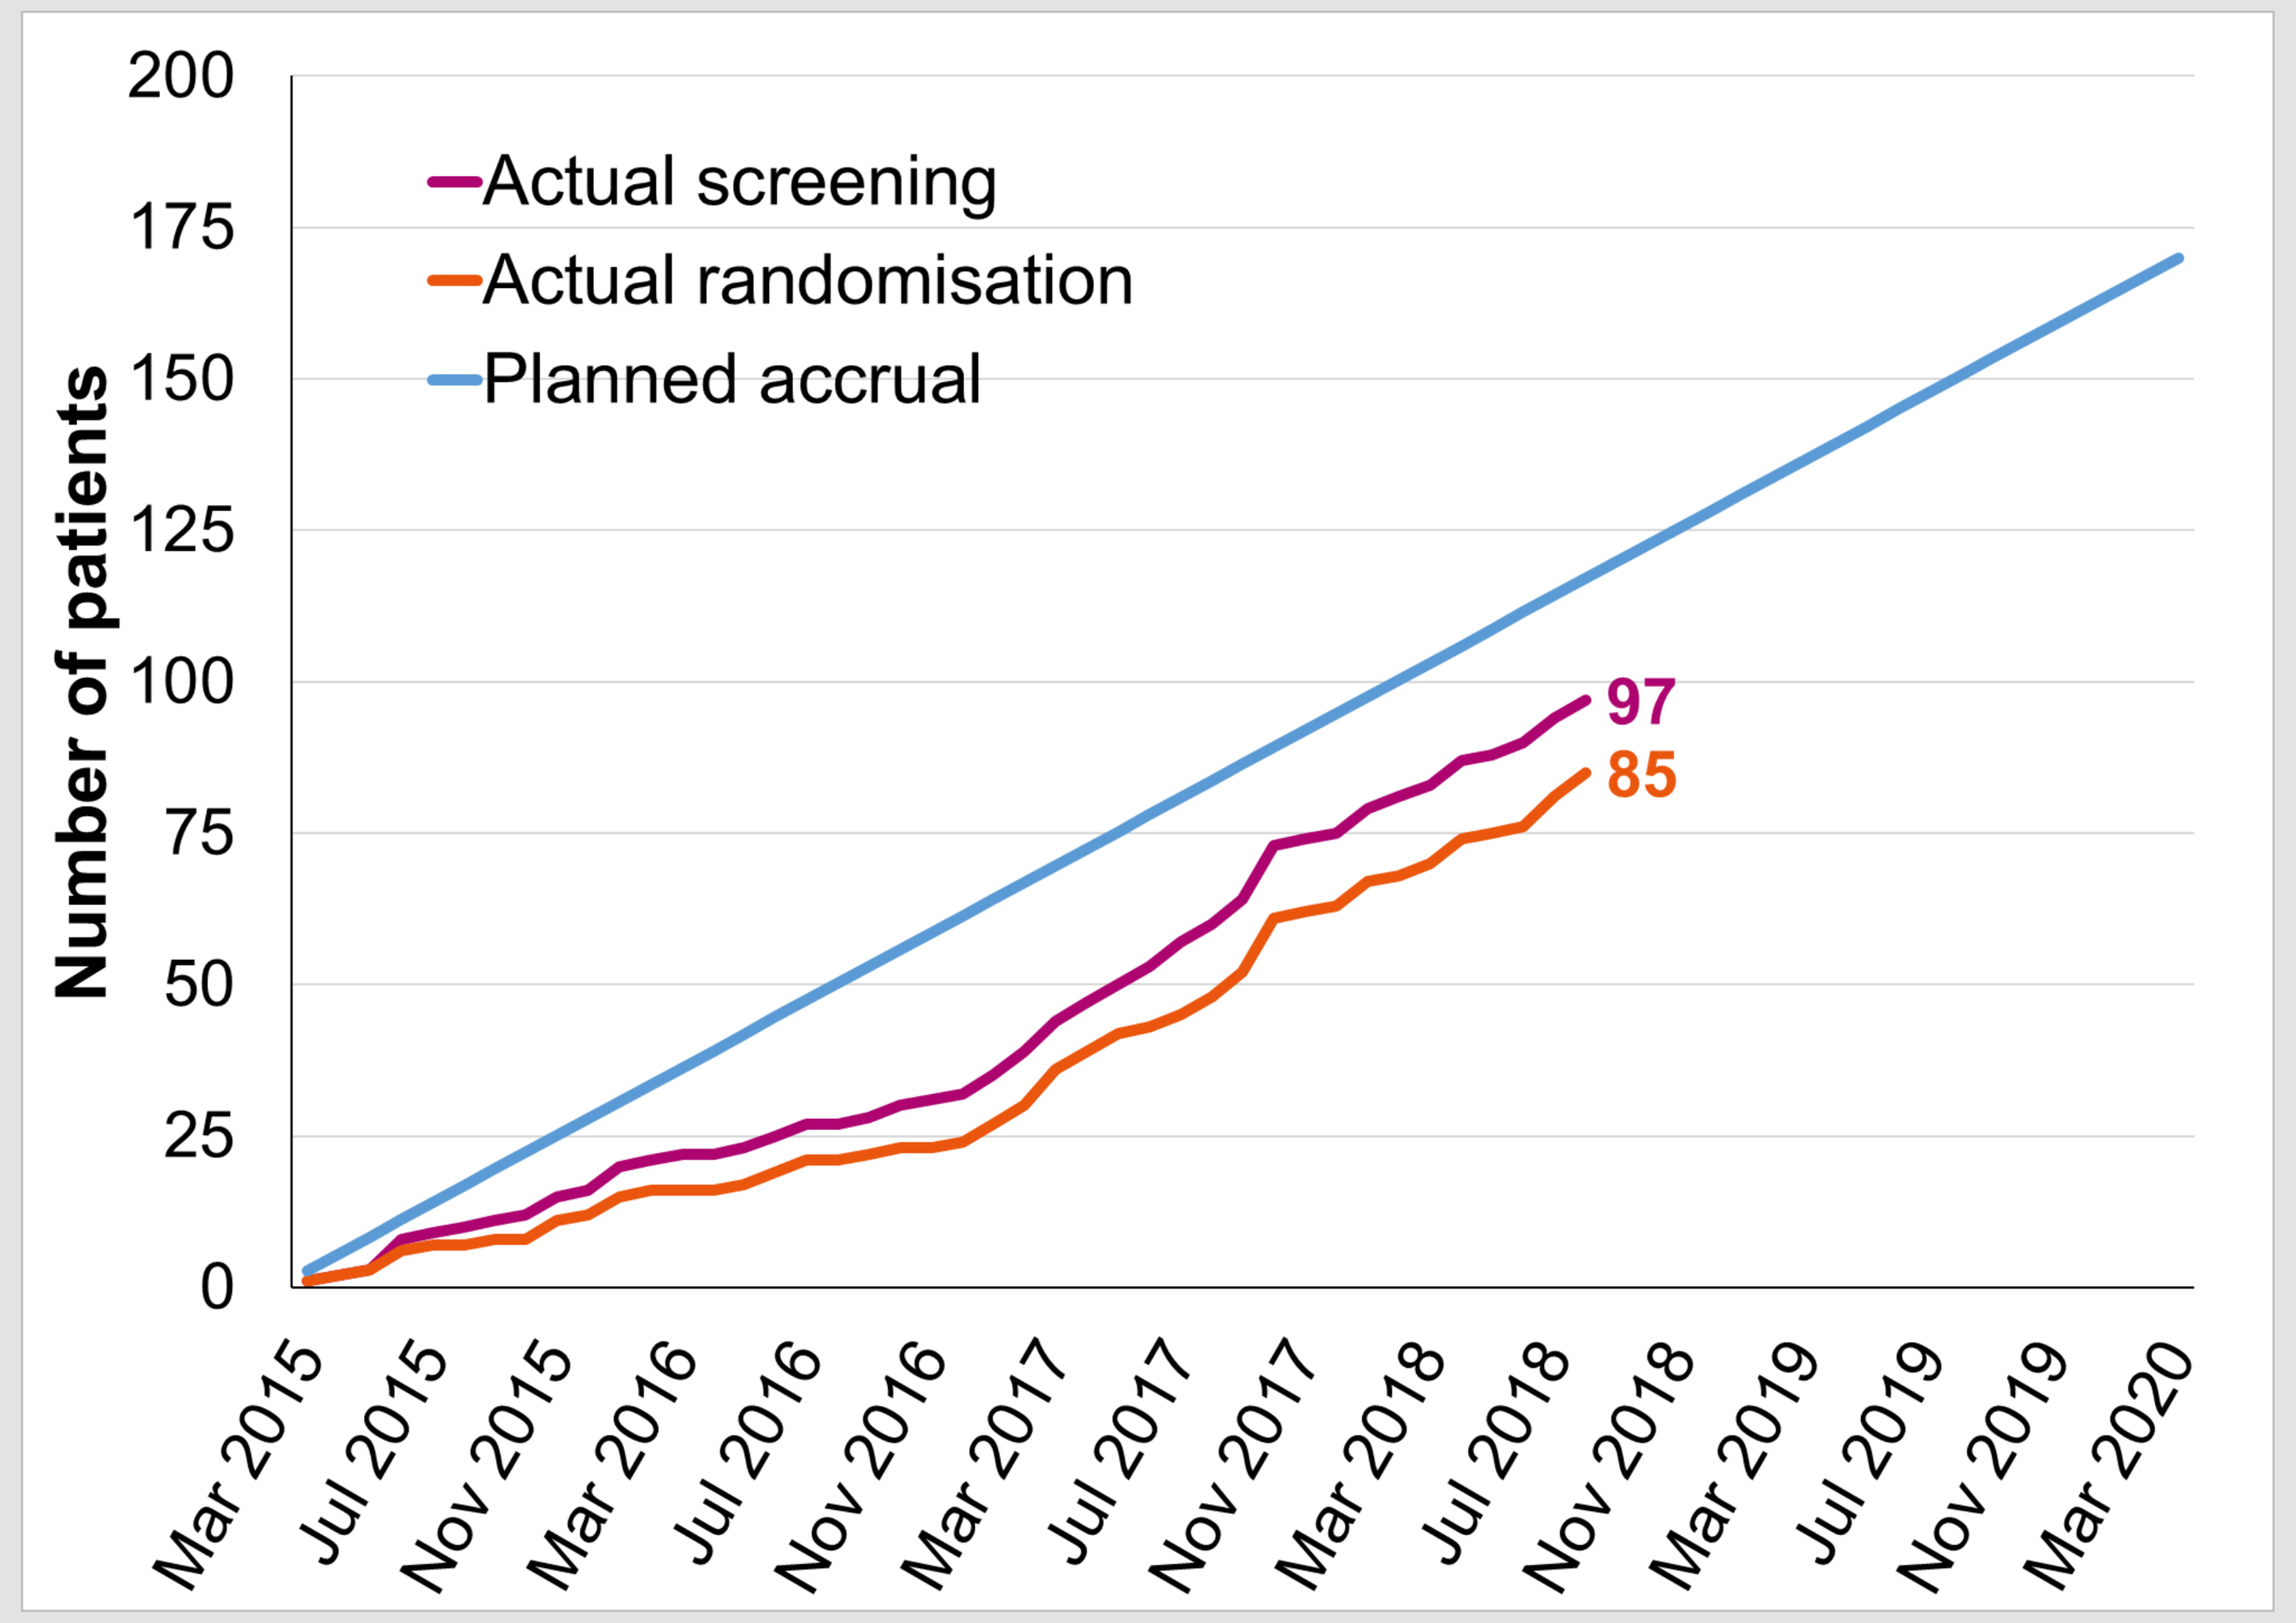
200
175
150
125
100
75
50
25
0
Number of patients
Actual screening
Actual randomisation
Planned accrual
97
85
Mar 2015
Jul 2015
Nov 2015
Mar 2016
Jul 2016
Nov 2016
Mar 2017
Jul 2017
Nov 2017
Mar 2018
Jul 2018
Nov 2018
Mar 2019
Jul 2019
Nov 2019
Mar 2020

Figure 3 - Accrual status

Study design

NEOPAN is a multicenter, phase 3, open-label randomised study (NCT02539537).

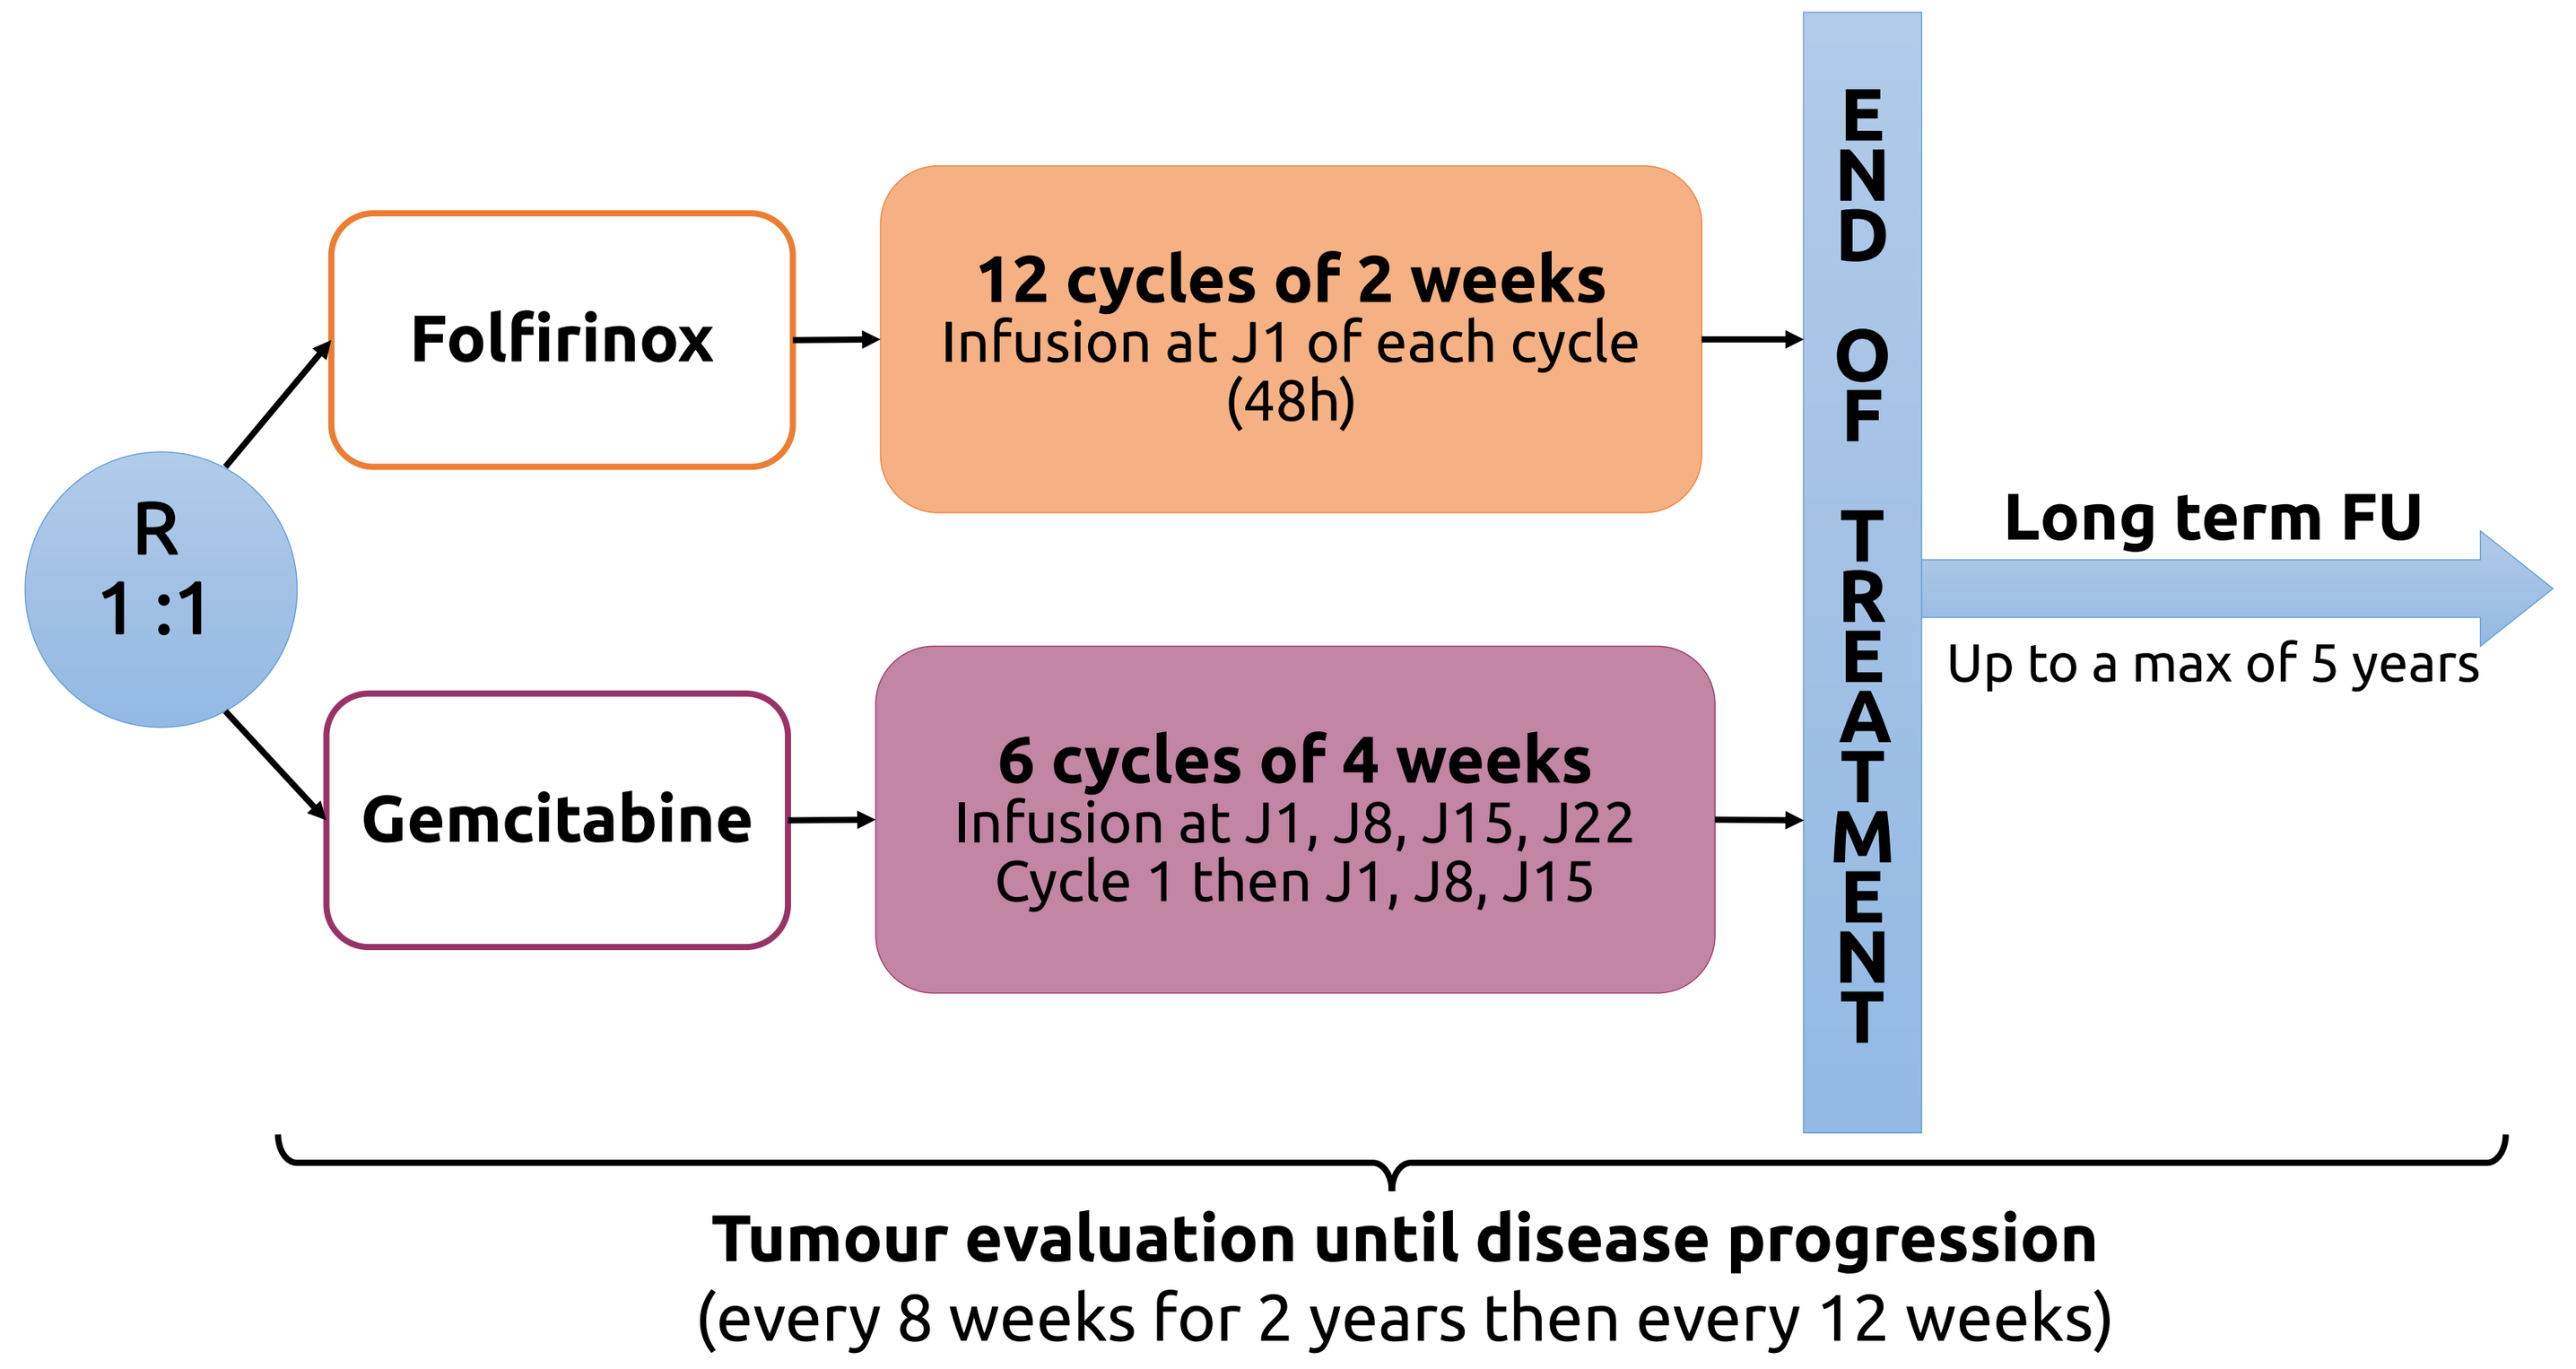
R
1 :1
Folfirinox
12 cycles of 2 weeks
Infusion at J1 of each cycle
(48h)
Gemcitabine
6 cycles of 4 weeks
Infusion at J1, J8, J15, J22
Cycle 1 then J1, J8, J15
END OF TREATMENT
Long term FU
Up to a max of 5 years
Tumour evaluation until disease progression
(every 8 weeks for 2 years then every 12 weeks)

Figure 1 - NEOPAN study design

Stratification factors for randomisation: Investigator site, Tumour location (head of the pancreas Yes vs. No), Performance status (ECOG 0 vs. 1), Age (≤ 60 years old vs. > 60 years old)

Patients treatment

Study treatment:

Patients will be treated as follows during 6 months:

- **Control arm:** Gemcitabine 1 000 mg/m^2, IV infusion over 30 min once per week, 3 weeks on/1 week off except for the first cycle with 4 infusions at D1, D8, D15, D22, repeated every 4 weeks for 6 cycles;
- **Experimental arm (mFolfirinox)**: leucovorin 400 mg/m^2 at D1, irinotecan 180 mg/m^2 at D1, oxaliplatin 85 mg/m^2 at D1, 5-FU 2400 mg/m^2 IV continuous infusion over 46h, no bolus of 5-FU, repeated every 2 weeks for 12 cycles.

Treatment recommendation after end of study treatment:

After this 6-month period, each center has to choose between 3 strategies: stop of treatment, maintenance chemotherapy with gemcitabine or capecitabine, radio-chemotherapy.

Statistical considerations

Sample size: 170 patients, 142 events, 5 years of accrual and follow-up of at least 6 months

Based on assumptions of PFS prolongation from 5 months (control arm) to 8 months (experimental arm), a-risk 5%(two-sided), power 80%

Interim analyses:

- 4 interim analyses planned for the composite index of induction treatment severe toxicity to control Folfirinox tolerance after 8 weeks of treatment (4 cycles)

Study Population

Main Inclusion Criteria

- Patients ≥ 18 year of age
- Cytologically or histologically proven adenocarcinoma of the pancreas
- Locally advanced PC proven to be unresectable after multidisciplinary discussion
- Performance status ECOG < 2
- Normal hematologic, hepatic, and renal functions
- Adequate other vital functions
- Measurable lesions (RECIST criteria)
- Signed informed consent

Main Exclusion Criteria

- Metastatic disease
- Treatment of another cancer within 5 years prior to recruitment, except for in situ cervical cancer and basal cell carcinoma
- Cardiac disorders grade III/IV according to NYHA
- Major comoribidty (HIV, chronic HBV or HBC, non controlled diabetis)
- Preexisting neuropathy grade ≥ 2, Gilbert disease or UGT1A1 *28/*28
- Pregnancy

Acknowledgment

We thank the patients and their families for participating in the study. We are also indebted to all the participating centers.

For additional information, please contact Prof. Michel DUCREUX , study coordinator: Michel.DUCREUX@gustaveroussy.fr