unicancer

FONDATION ARC POUR LA RECHERCHE SUR LE CANCER
Reconnue d'utilité publique

EXPRESS Study - A Multicenter, Prospective Trial In Progress Exploring The Association Between Low Level Of Genomic Alteration And Exceptional And Unexpected Response To Targeted Therapies In Patients With Solid Tumours

A.Italiano[1], O. Le Saux[2], F. André[3], T. Filleron[4], D. Spaeth[5], P.-E. Heudel[6], L. Albiges[3], T. Bachelot[6], A. Gonçalves[7], J.-Y. Pierga[8], F. Barlesi[9], V. Boige[6], C. Lebbe[10], L. Mortier[11], J.-S. Frenel[12], O. Tredan[6], M. Jimenez[13], F. Legrand[13], C. Ferté[3];

[1]Institut Bergonié, Bordeaux, France, [2]Centre Hospitalier Lyon Sud - Hospices Civils de Lyon, Pierre Benite, France; [3]Gustave Roussy, Villejuif, France; [4]Institut Universitaire du Cancer de Toulouse, Toulouse, France; [5]Centre d'Oncologie de Gentilly, Nancy, France; [6]Centre Léon Bérard, Lyon, France; [7]Centre Paoli Calmette, Marseille, France; [8]Institut Curie, Paris, France; [9]AP-HM, Marseille, France; [10]Hôpital Saint-Louis, Paris, France; [11]CHU de Lille, Lille, France; [12]ICO- Centre René Gauducheau, Nantes, France; [13]UNICANCER R&D, Paris, France;

EXPRESS exceptional response

MUNICH 2018 ESMO congress

Background

Most targeted therapies in cancer have reached approval based on clinical studies performed in unselected patients. Molecular anomalies predictive of the response to targeted therapies remain largely unknown.

Small subsets of patients treated with targeted therapy present exceptional and unexpected response (1-10%), which could be driven and correlated by a low level of genomic alterations in genes identified as causally implicated in cancer.

Conversely, the multiplication of genomic alterations in such genes would decrease the oncogenic de-addiction process (1, 2).

References

1- Fu Y, Cancer Pharmacogenomics and Targeted Therapies conference 2014; Cambridge, UK

2- de Bruin EC, McGranahan N, Mitter R et al. Spatial and temporal diversity in genomic instability processes defines lung cancer evolution. Science. 2014 Oct 10;346(6206) -251-6. doi - 10.1126/science.1253462.

Objectives

Primary objective :

- **To identify whether tumours characterised by a low level of genomic alteration in genes** identified as causally implicated in cancer are associated with exceptional and unexpected response across anticancer targeted therapies.

Secondary objectives :

- **To compare the rate of tumours with a low level of genomic alterations** between the EXPRESS cohort and the control cohort of patients.
- **To identify novel candidate somatic molecular profiles** susceptible to drive exceptional and unexpected response across anticancer targeted therapies.
- To identify novel candidate somatic molecular profiles associated with exceptional response within given classes of anticancer targeted therapies.
- **To identify host immunologic** characteristics associated with exceptional response.
- **To guide further exploratory and confirmatory studies** (i.e. functional validation) based on the clinical and genomic data and biological resources raised from EXPRESS project.

Participating Centres

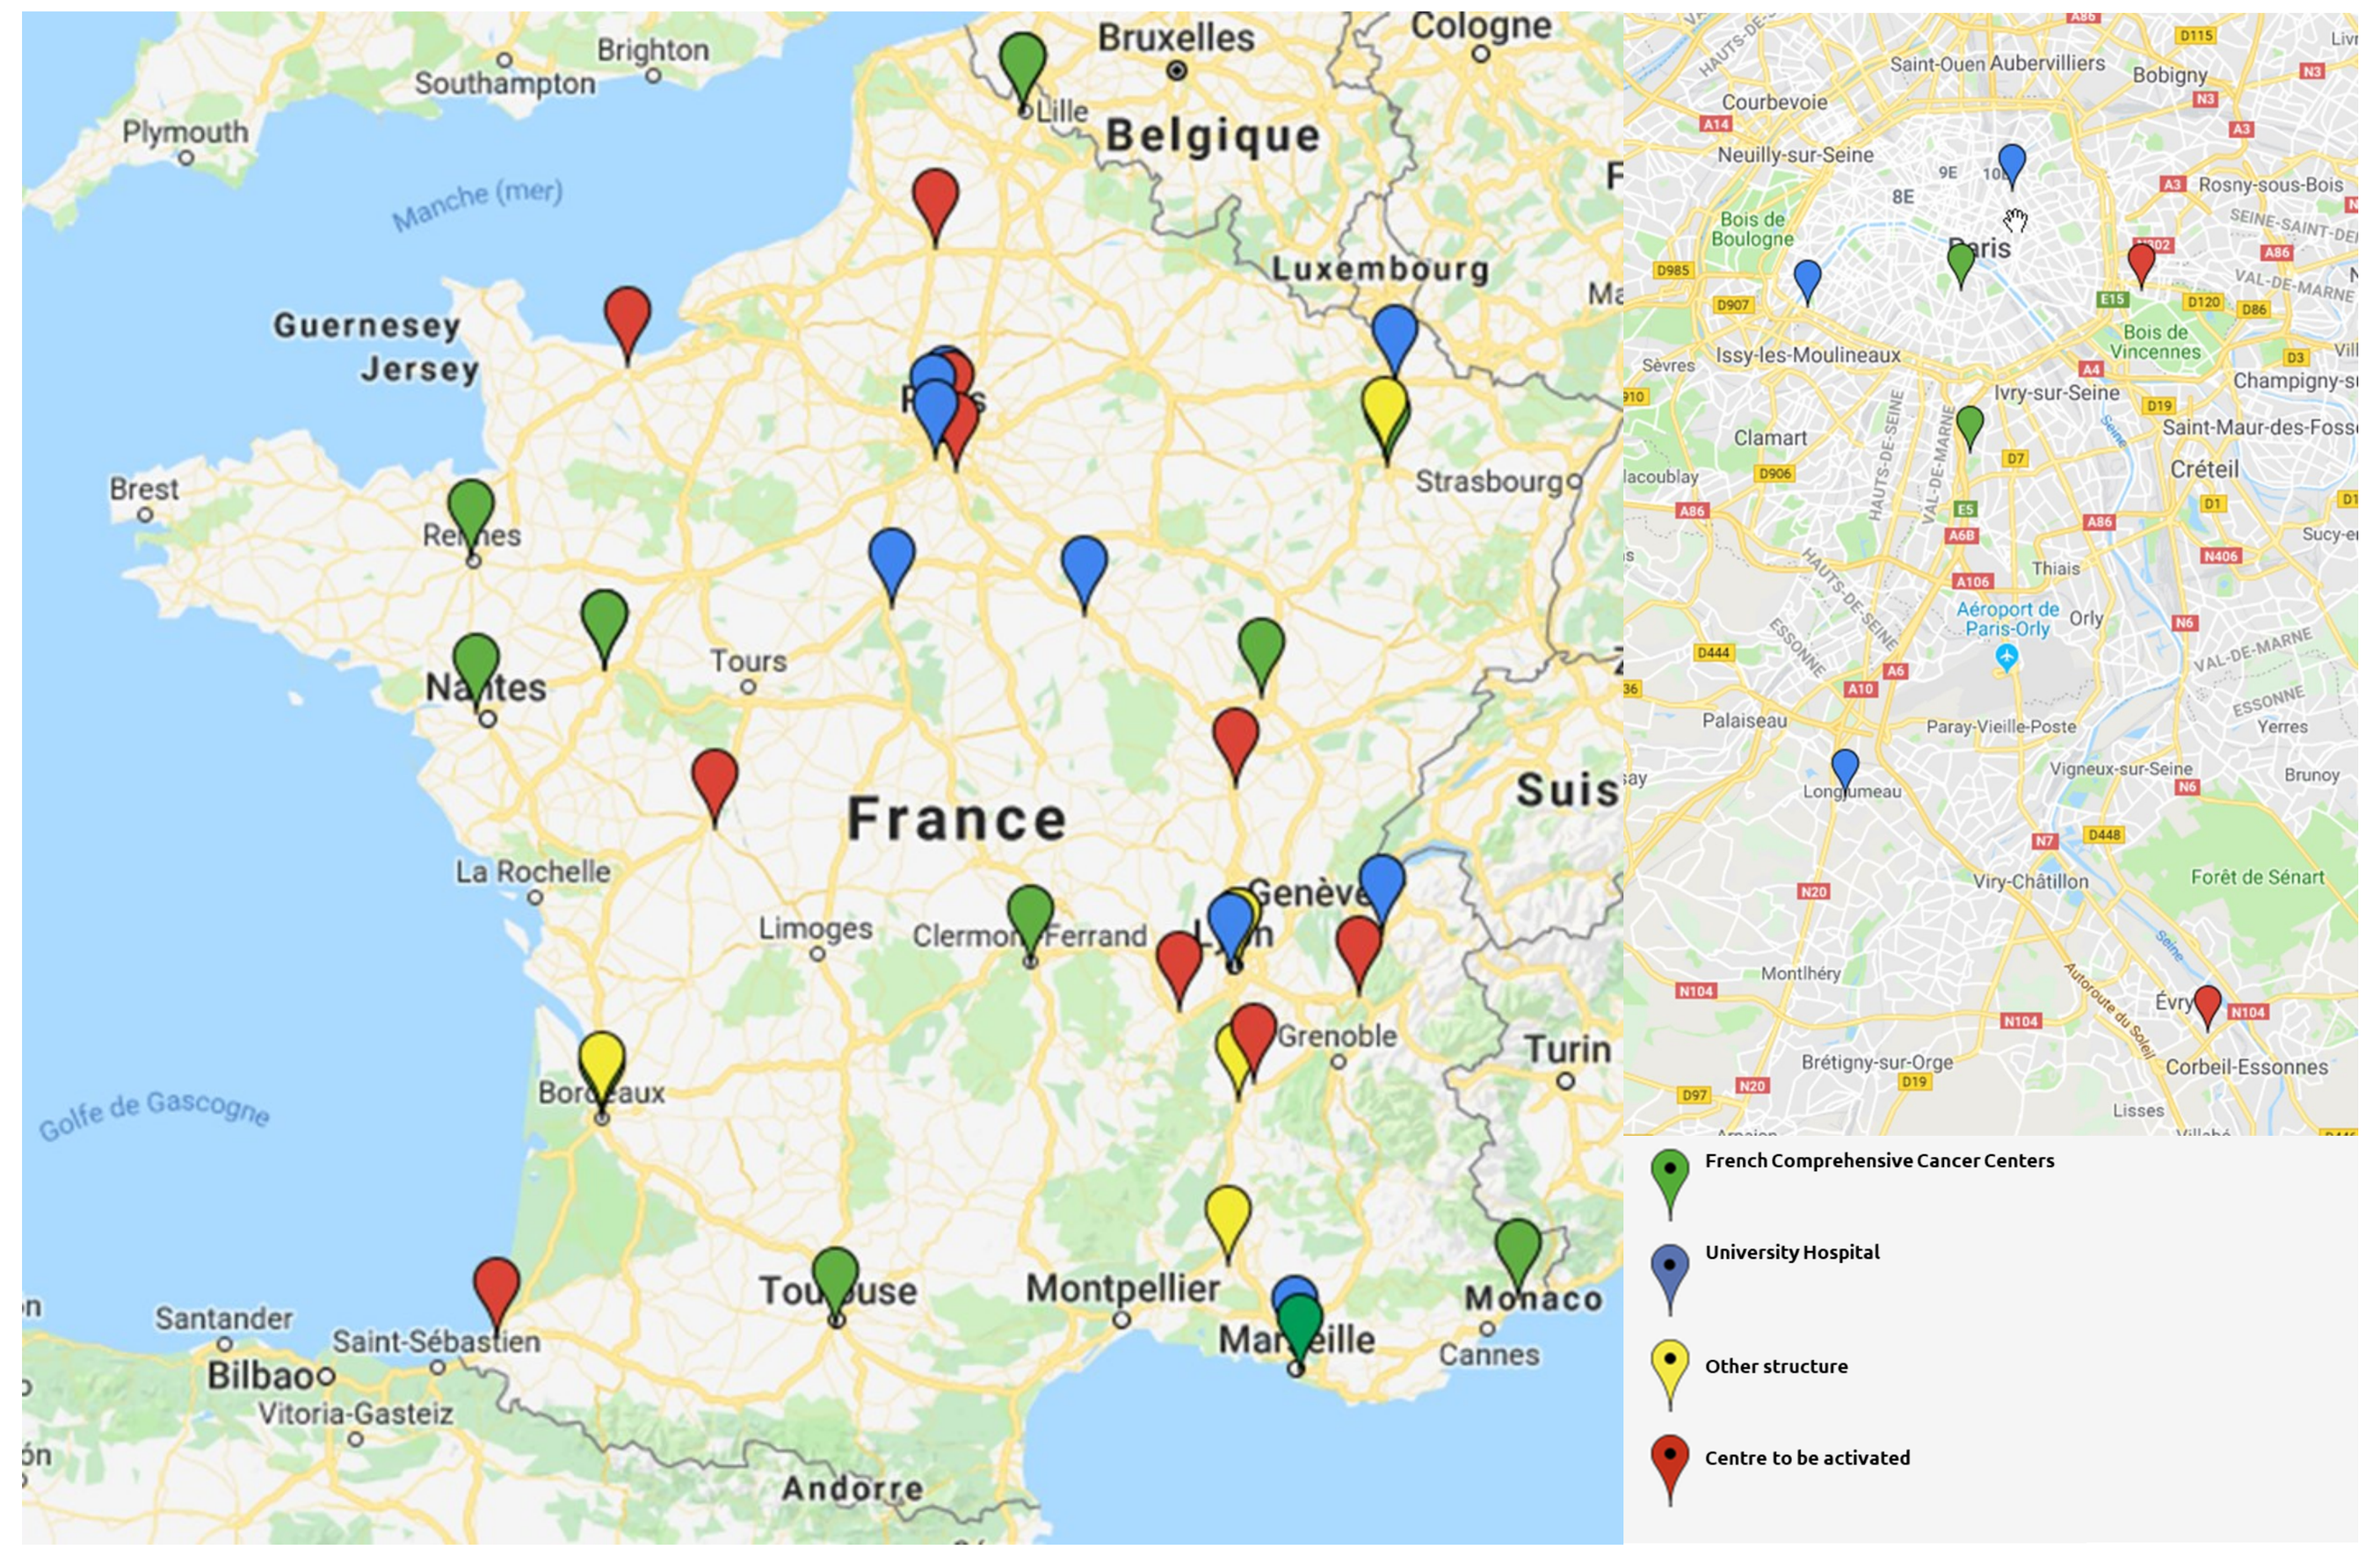
French Comprehensive Cancer Centers
University Hospital
Other structure
Centre to be activated

Figure 2 - Map of centres already involved in the study

Methodology

Exceptional tumour responses are naturally rare and unexpected; patients will be identified retrospectively and in a nationwide manner (figure 1). Only this unique approach enables the recruitment of a substantial number of exceptional responders in a relatively short period of time.

The definition of **exceptional and unexpected response** to an approved antineoplastic targeted therapy in patients with advanced solid cancers (breast, lung, colorectal, ovarian, kidney or melanoma) is of paramount importance and will be confirmed monthly by the Response Confirmation Committee (COREV) composed of the study coordinators and at least one expert of each organ.

We decided to apply the **definition chosen by the NCI**, which combines three criteria:

1. **A complete response or a partial response (RECIST),**

AND

2. **Lasting more than 6 months,**

AND

3. **Not expected in more than 10% of the patients in this drug – organ type situation.**

For each tumour type, we wish to test the null hypothesis H0 - $\pi=0.05$ against the one-sided alternative hypothesis $\pi>0.05$. For each of the six cohorts, a sample size of 44 patients is necessary to achieve 80% power at $\pi=15$ with a one-sided level 5% test.

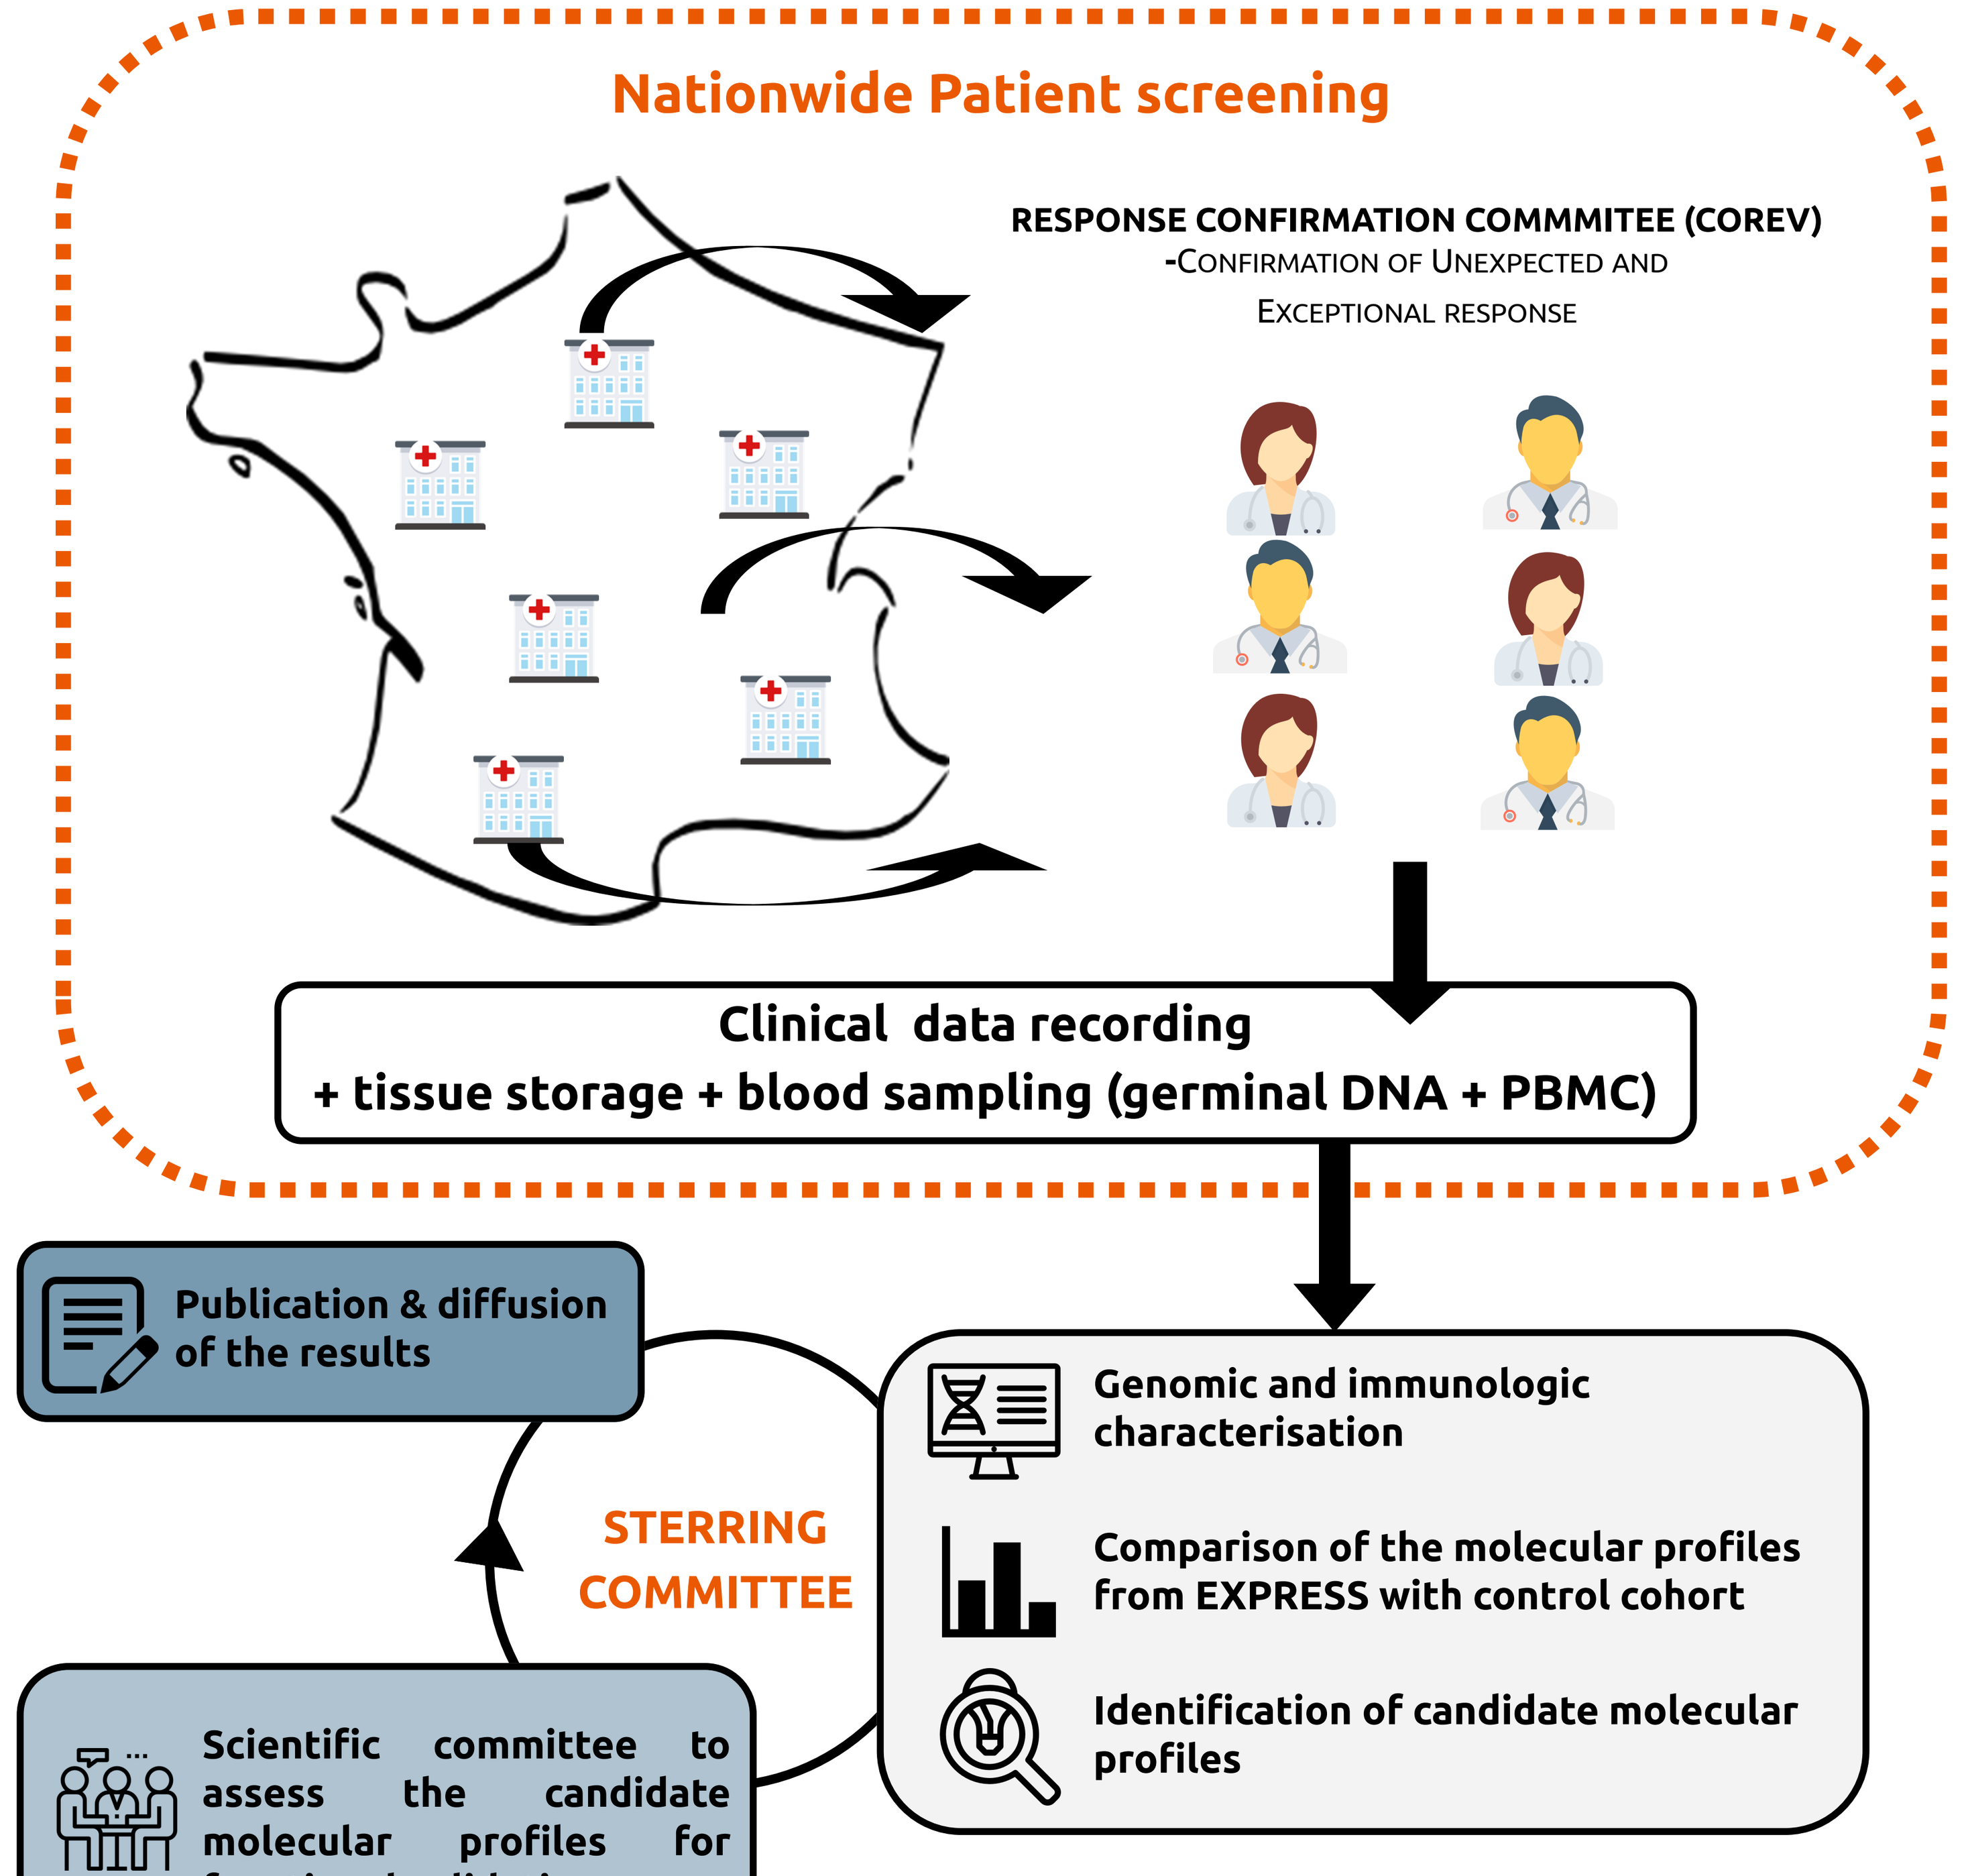
Nationwide Patient screening
RESPONSE CONFIRMATION COMMMITEE (COREV)
-Confirmation of Unexpected and Exceptional response
Clinical data recording
+ tissue storage + blood sampling (germinal DNA + PBMC)
Publication & diffusion of the results
STERRING COMMITTEE
Genomic and immunologic characterisation
Comparison of the molecular profiles from EXPRESS with control cohort
Identification of candidate molecular profiles
Scientific committee to assess the candidate molecular profiles for functional validation

Figure 1 - Study workflow

Main Inclusion Criteria

- Adult patient (≥18 years old at diagnosis).
- Patient suffering from the following tumour type - breast cancer, lung adenocarcinoma or squamous cell carcinoma, colorectal cancer, ovarian cancer, renal clear cell cancer, skin cutaneous melanoma.
- Metastatic or locally advanced disease.
- Currently or previously treated with an anticancer targeted therapy in monotherapy. Targeted therapies combined with other agents are accepted only if:
 - the tumour was previously proven to be progressive under the same agents,

 or

 - the response or the stability has been maintained with the targeted therapy alone after the agent has been stopped.
- Exceptional and unexpected tumour response to any marketed targeted therapy confirmed by the college of experts.
- Availability and required quality of the tumour biopsy (FFPE or frozen sample) allowing for the whole exome sequencing analysis. Tumour biopsies obtained just before the initiation of the targeted therapy are preferred; otherwise any prior sample is possible.

Recruitment (September 2018)

As of September 2018, **203** patients were identified over 37 centres (figure 2). Among them **95** were potentially incluable, and **63** were already included among the 6 cohorts (figure 3).

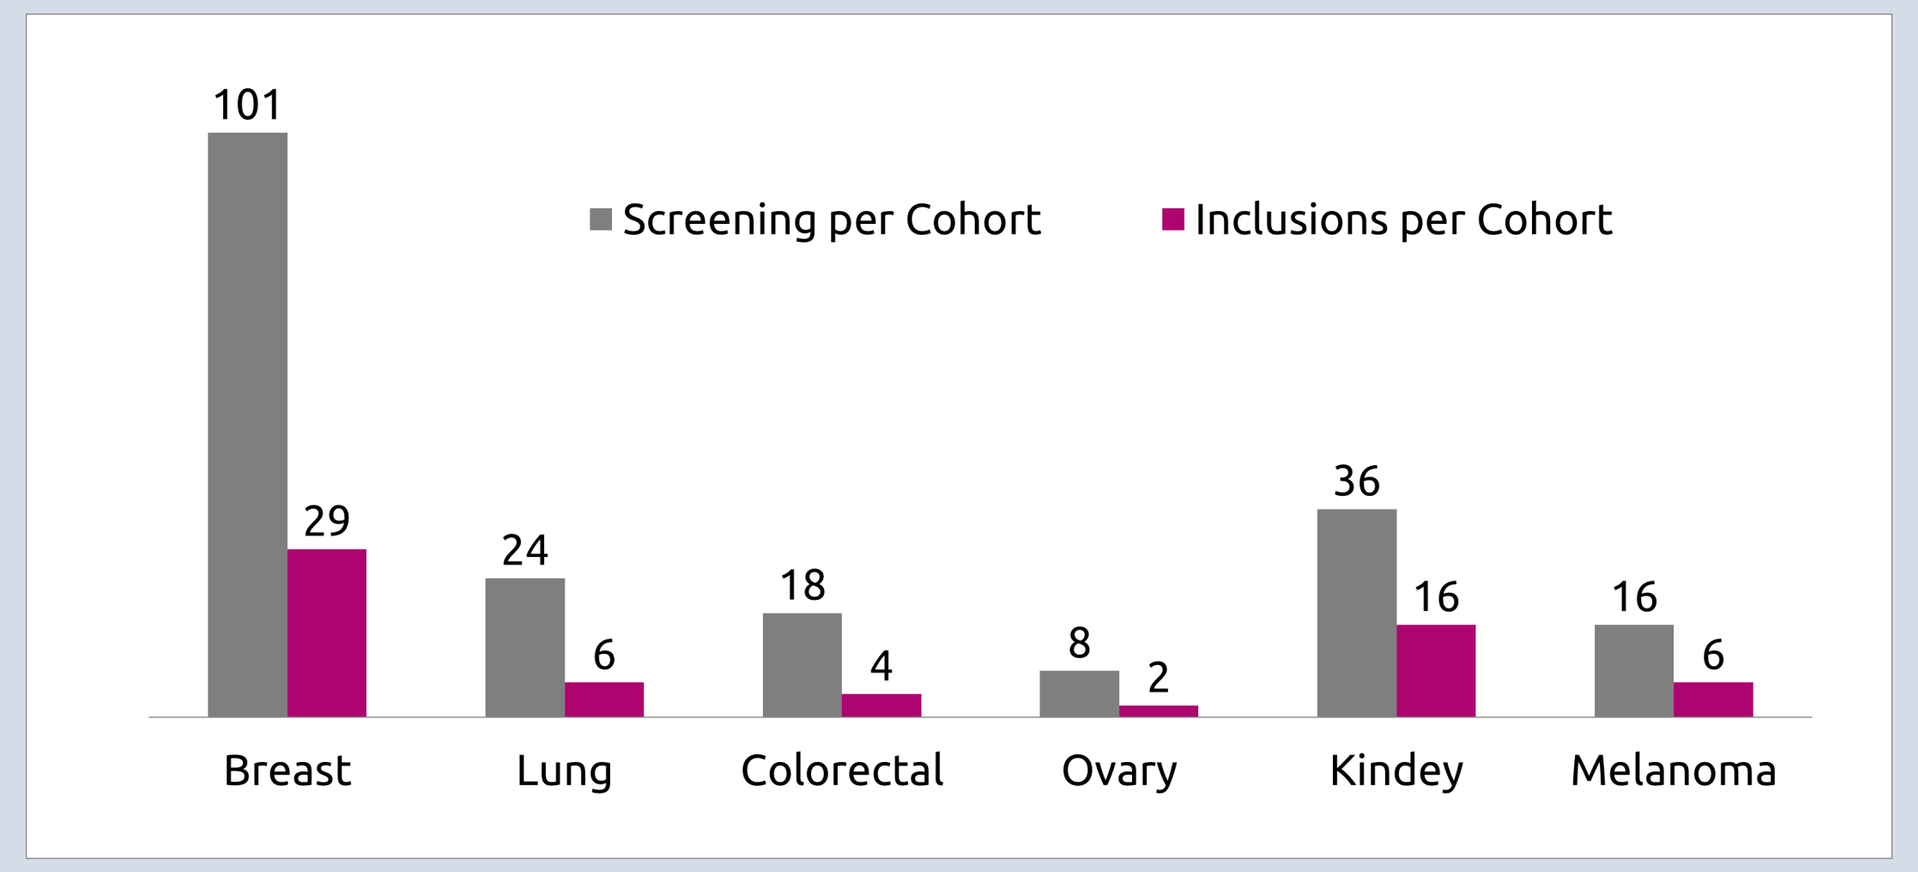
Screening per Cohort
Inclusions per Cohort
101
29
24
6
18
4
8
2
36
16
16
6
Breast
Lung
Colorectal
Ovary
Kindey
Melanoma

Figure 3 - Screening and Inclusion per cohort

Breast cancer cohort is half full (29/44), and 16 patients are already included in the kidney cohort. For other cohorts, identification of potential patients is challenging due to the limited number of targeted therapy available in these indication, and the growing interest in immunotherapy.

Figure 4 shows the variety of targeted therapy received by patients included in the study.

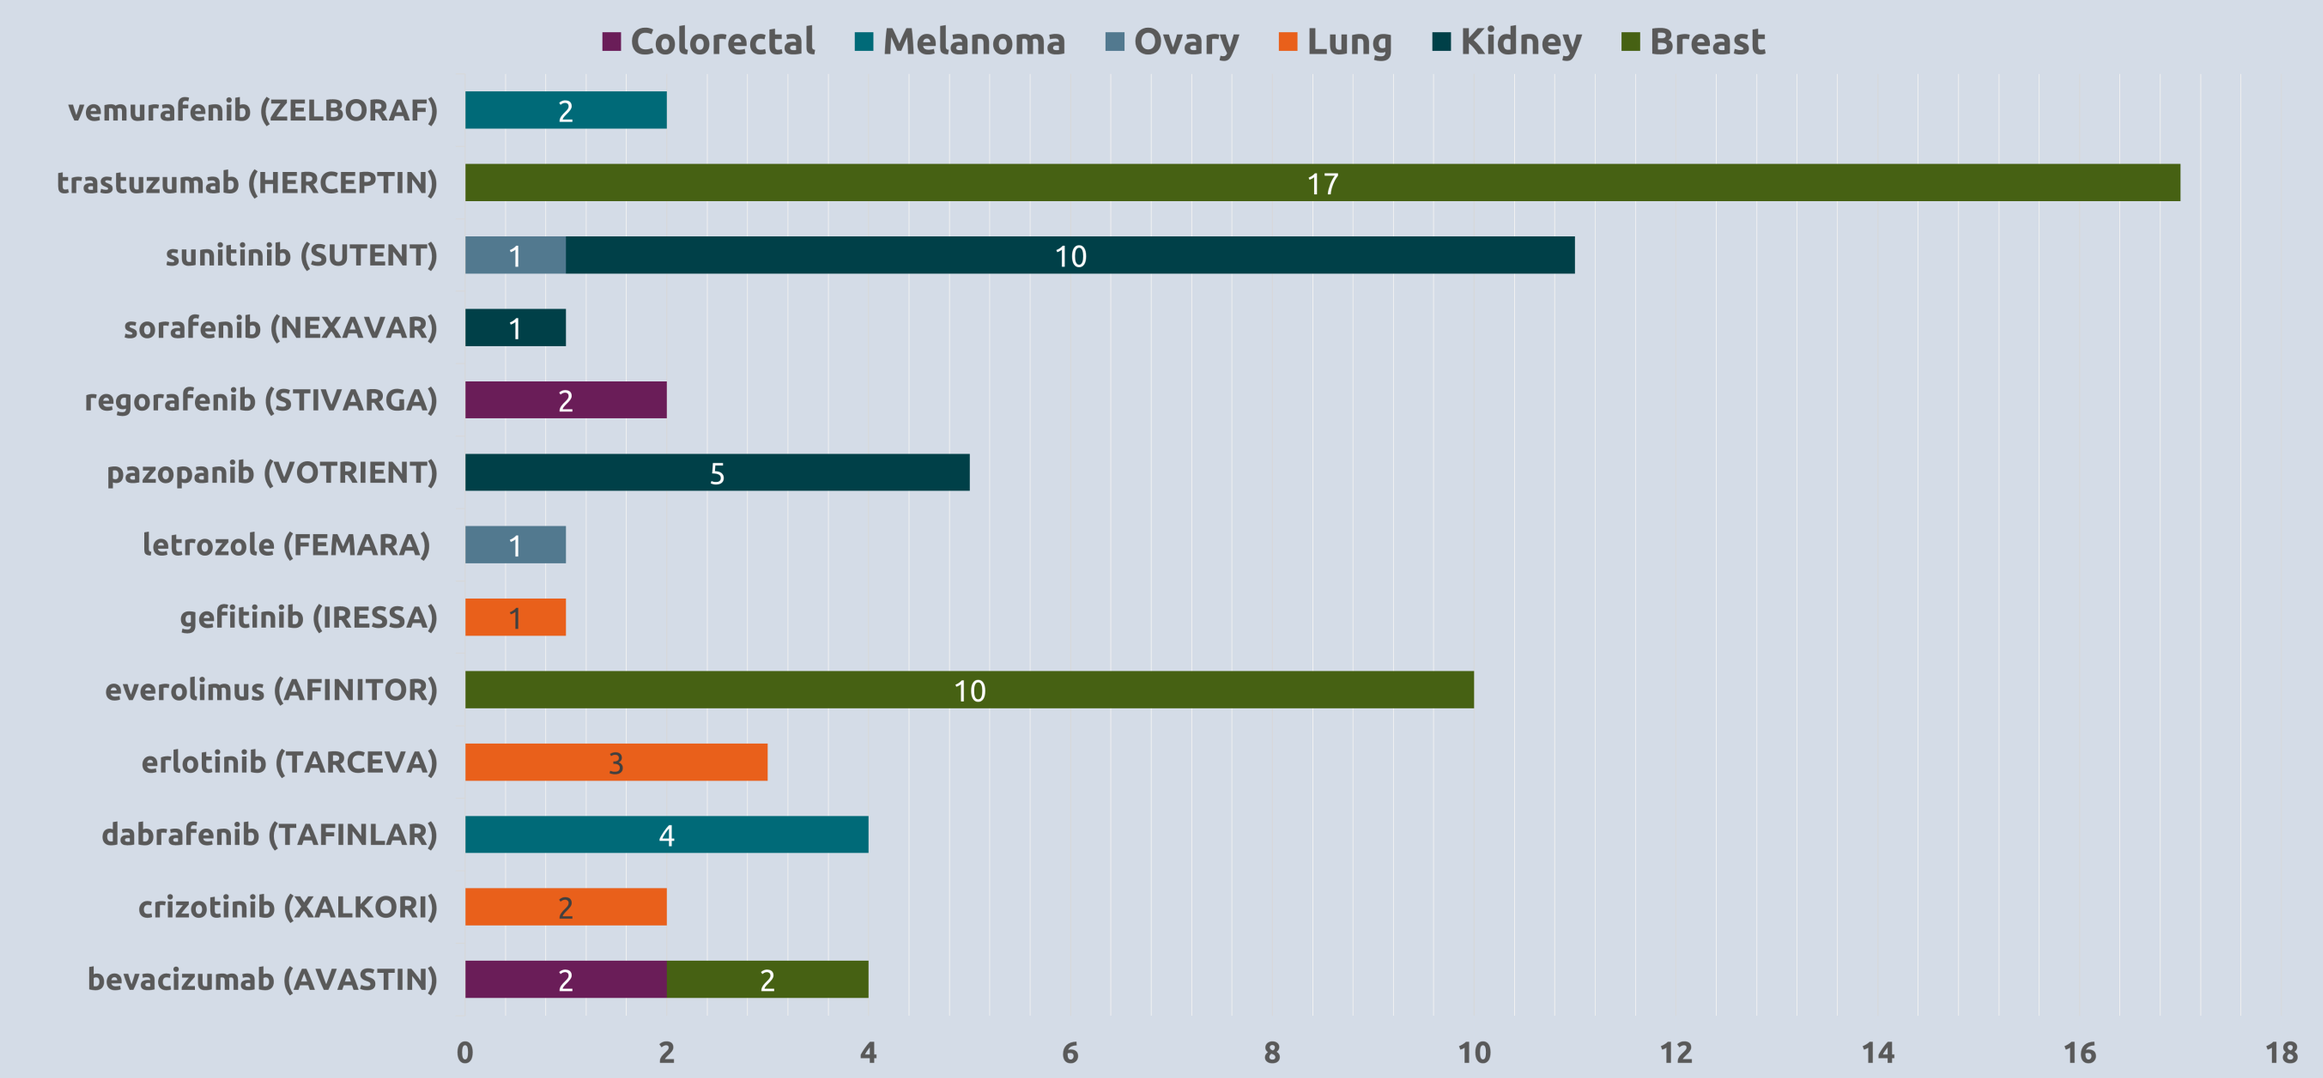
Colorectal
Melanoma
Ovary
Lung
Kidney
Breast
vemurafenib (ZELBORAF) 2
trastuzumab (HERCEPTIN) 17
sunitinib (SUTENT) 1 10
sorafenib (NEXAVAR) 1
regorafenib (STIVARGA) 2
pazopanib (VOTRIENT) 5
letrozole (FEMARA) 1
gefitinib (IRESSA) 1
everolimus (AFINITOR) 10
erlotinib (TARCEVA) 3
dabrafenib (TAFINLAR) 4
crizotinib (XALKORI) 2
bevacizumab (AVASTIN) 2 2
0
2
4
6
8
10
12
14
16
18

Figure 4 - Targeted therapy received by included patient

Conclusion

The identification of molecular traits associated with exceptional responder might serve the development of predictive classifiers for precision medicine. This study also represents a unique opportunity to better understand cancer biology. **The study is still recruiting**, completion date is estimated to be in August 2019. Other tumour cases were sometimes sent and collected, and the opening of additional cohorts is **under discussion.**

Acknowledgment

Patients and families , Financial support of the ARC Foundation, Dr. Charles Ferté

Additional information is available on Unicancer website, including patient information and screening form : http -//www.unicancer.fr/rd-unicancer/etude-express