434P

Abstract #5138

2018 ESMO congress, MUNICH, GERMANY

Biomarker-driven access to crizotinib in ALK, MET or ROS1 positive $^{(+)}$ malignancies in adults and children: the French national AcSé Program.

G. Vassal[1], Nathalie Cozic[2], Roch Houot[3], Laurence Brugières[4], Thomas Aparicio[5], Jean-Yves Blay[6], Maurice Pérol[6], Pauline Brice[7], Emeline Meriaux[8], Birgit Geoerger[4], Marie-Rose El Bejjani[9], Salma Moalla[9], Ivan Bièche[10], Sylvie Lantuejoul[11], Céline Mahier Ait Oukhatar[12], Natalie Hoog Labouret[13], Denis Moro-Sibilot[14].

1 - Clinical Research Department, Gustave Roussy Cancer Campus, Villejuif, FR ; 2 - Oncostat, Gustave Roussy Cancer Campus, Villejuif, FR ; 3 - Hematology, Hôpital Pontchaillou, Rennes, FR ; 4 - Department of Pediatric and Adolescent Oncology, Gustave Roussy Cancer Campus Grand Paris, Villejuif, FR ;
5 - Gastroenterology and Digestive Oncology, Hôpital Saint Louis, APHP, Paris, FR ; 6 - Medical Oncology, Centre Léon Bérard, Lyon, FR ; 7 - Hematology and oncology, Hôpital St. Louis, APHP, Paris, FR ; 8 - Medical Oncology, ICO Institut de Cancérologie de l'Ouest René Gauducheau, Saint-Herblain, FR ; 9 - Radiology, Gustave Roussy Cancer Campus, Villejuif, FR ;
10 - Pharmacogenomics Unit, Institut Curie, Paris, FR ; 11 - Department of Biopathology, Centre Léon Bérard, Lyon, FR ; 12 - Research and Development, UNICANCER, Paris, FR ; 13 - Biology, Transfert and Innovation Department, Institut National du Cancer, Boulogne Billancourt, FR ; 14 - Thoracic Pneumology, Hôpital Michallon, Grenoble, FR.

AcSé Program : secured access program to innovative cancer drugs

Background

- When a marketed targeted therapy exists in a molecularly defined subgroup of patients
- When the same alteration is found in other tumor types

Risk of a wide off label use of the drug

Objectives

1/ Promote a secured access for all patients with an advanced refractory malignancy and no therapeutical alternative through an academic phase II clinical trial.

- One trial for each targeted treatment selected
- Withdrawal if high toxicity or no efficacy in a predefined number of patients with the same tumor type
- Efficacy signal force to inform pharmaceutical firms for drug development decision making

2/ Ensuring equity of access to innovation

- Provide nationwide molecular tumor diagnosis for all patients through INCa molecular genetic centers
- Whatever the healthcare institution status (public hospitals, private hospitals…)
- Perform high quality tests
- Hemopathies, solid tumors

France organization of the 28 molecular centers for personalized medicine

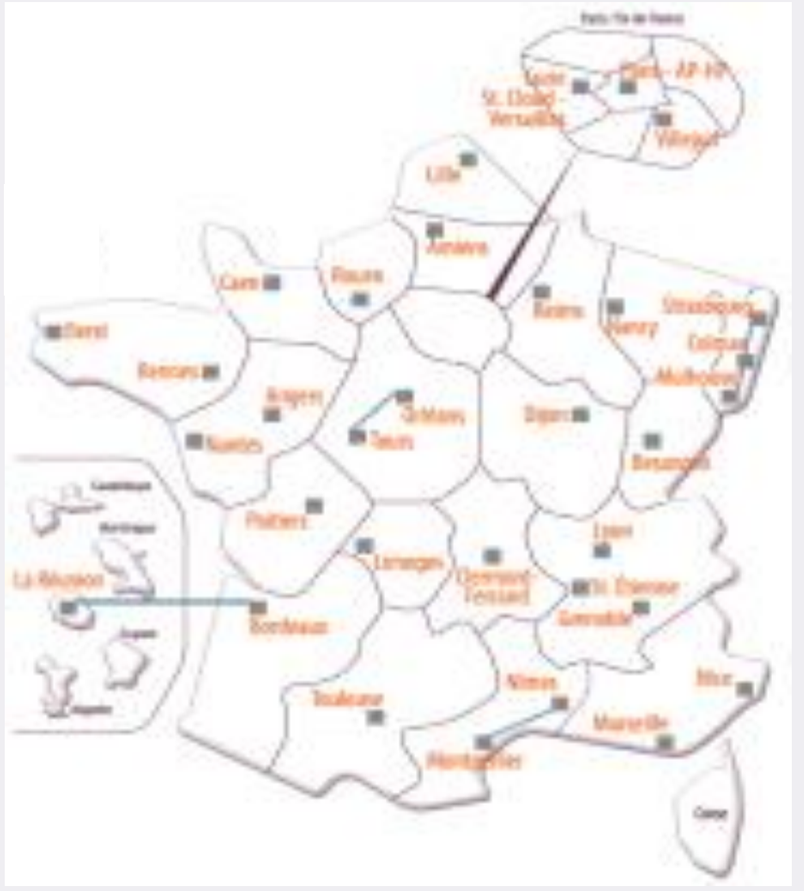

Partnerships between laboratories located in University Hospitals and Cancer Centers

- Regional organization
- Cooperation between pathologists and biologists AcSé program

AcSé program

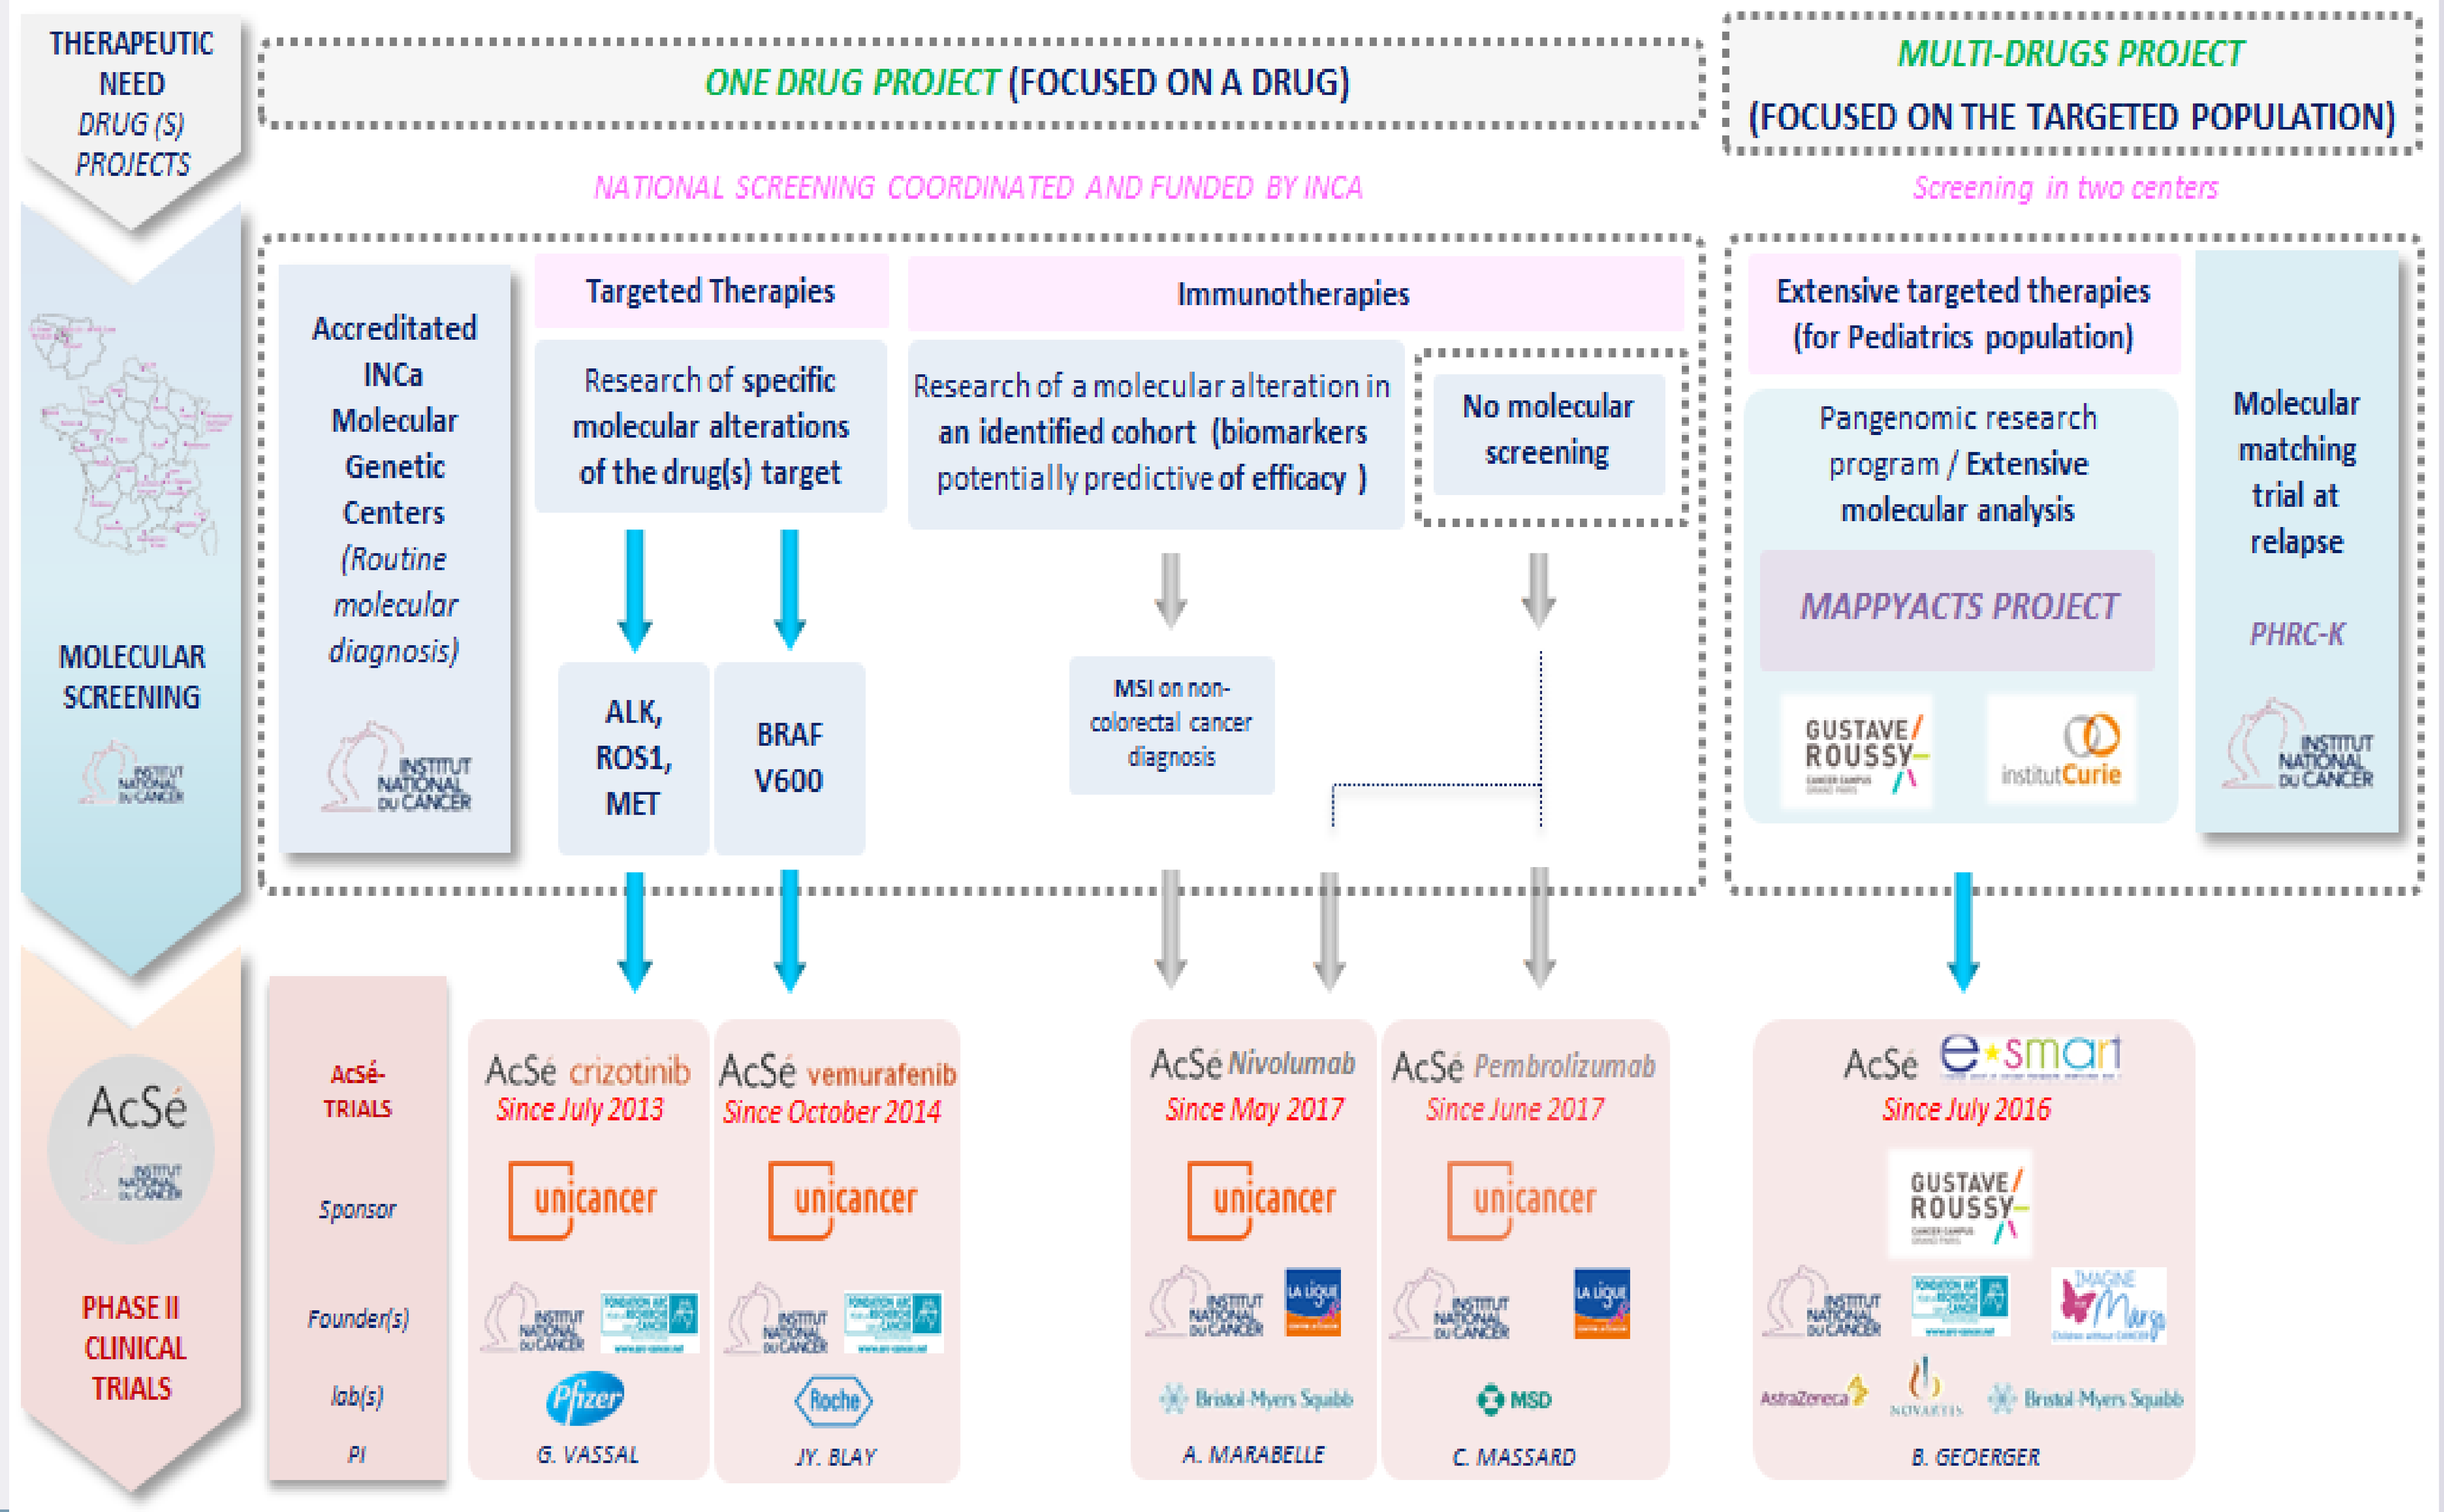

THERAPEUTIC NEED DRUG (S) PROJECTS
ONE DRUG PROJECT (FOCUSED ON A DRUG)
MULTI-DRUGS PROJECT (FOCUSED ON THE TARGETED POPULATION)
NATIONAL SCREENING COORDINATED AND FUNDED BY INCA
Screening in two centers
MOLECULAR SCREENING
Accreditated INCa Molecular Genetic Centers (Routine molecular diagnosis)
Targeted Therapies
Research of specific molecular alterations of the drug(s) target
ALK, ROS1, MET
BRAF V600
Immunotherapies
Research of a molecular alteration in an identified cohort (biomarkers potentially predictive of efficacy)
No molecular screening
MSI on non-colorectal cancer diagnosis
Extensive targeted therapies (for Pediatrics population)
Pangenomic research program / Extensive molecular analysis
MAPPYACTS PROJECT
Molecular matching trial at relapse
PHRC-K
PHASE II CLINICAL TRIALS
AcSé-TRIALS
Sponsor
Founder(s)
lab(s)
PI
AcSé crizotinib Since July 2013
G. VASSAL
AcSé vemurafenib Since October 2014
JY. BLAY
AcSé Nivolumab Since May 2017
A. MARABELLE
AcSé Pembrolizumab Since June 2017
C. MASSARD
AcSé e-smart Since July 2016
B. GEOERGER

AcSé crizotinib

Phase 2 clinical trial: Secured Access to Crizotinib for patients with tumors harboring a genomic alteration on one of the biological targets of the drug – PI : Gilles VASSAL / NCT02034981

Background

- Crizotinib is registered for treating ALK and ROS1-translocated lung cancer.
- Crizotinib is also a MET inhibitor.
- Crizotinib activity in MET-amplified (+) tumors was explored within the French National Cancer Institute (INCa) AcSé program.
- This included access to tumor molecular diagnoses and an exploratory multi-tumor 2-stage design phase II trial.

Main objective

Identification of subsets of patients that may benefit from treatment

Population

- Patients ≥ 1 year.
- Advanced disease harboring a genomic alteration in a crizotinib target.
- Patients not eligible for any other active academic or industry trial targeting the same alteration.

Molecular diagnosis process

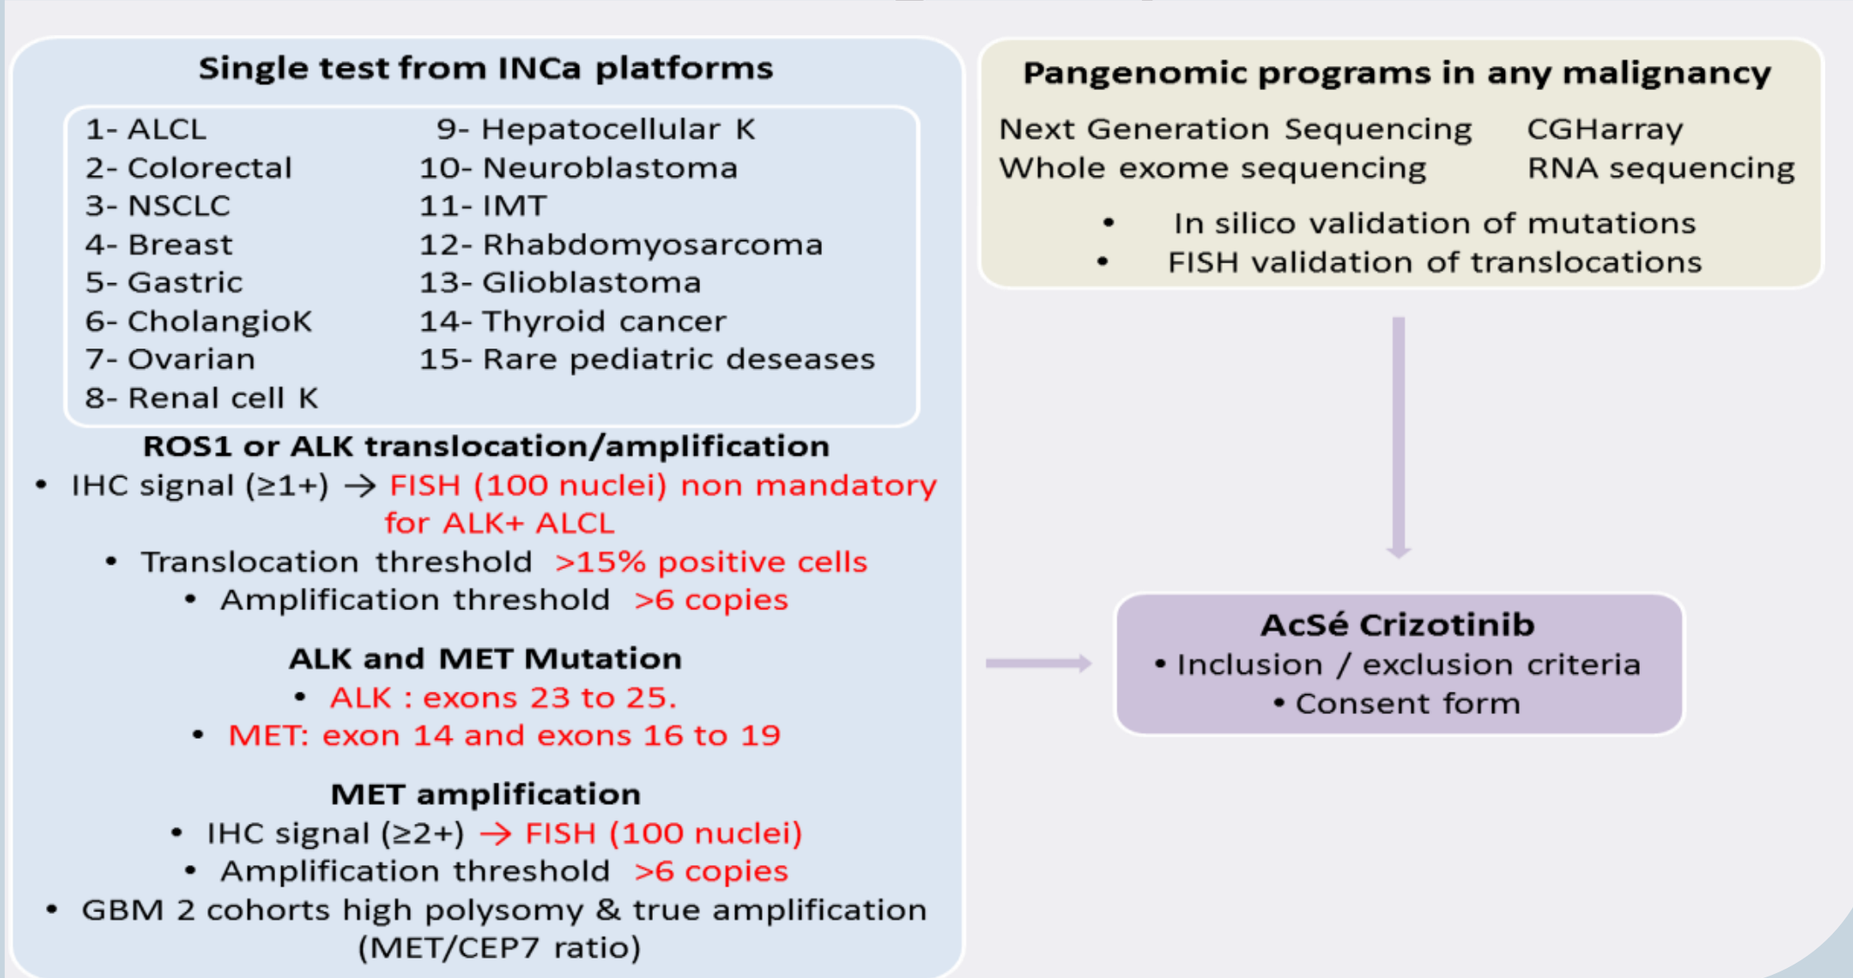

Single test from INCa platforms
1- ALCL
2- Colorectal
3- NSCLC
4- Breast
5- Gastric
6- CholangioK
7- Ovarian
8- Renal cell K
9- Hepatocellular K
10- Neuroblastoma
11- IMT
12- Rhabdomyosarcoma
13- Glioblastoma
14- Thyroid cancer
15- Rare pediatric deseases
ROS1 or ALK translocation/amplification
IHC signal (≥1+) → FISH (100 nuclei) non mandatory for ALK+ ALCL
Translocation threshold >15% positive cells
Amplification threshold >6 copies
ALK and MET Mutation
ALK : exons 23 to 25.
MET: exon 14 and exons 16 to 19
MET amplification
IHC signal (≥2+) → FISH (100 nuclei)
Amplification threshold >6 copies
GBM 2 cohorts high polysomy & true amplification (MET/CEP7 ratio)
Pangenomic programs in any malignancy
Next Generation Sequencing
Whole exome sequencing
CGHarray
RNA sequencing
In silico validation of mutations
FISH validation of translocations
AcSé Crizotinib
Inclusion / exclusion criteria
Consent form

Biomarker testing

Whole screening activity (at March, 2018)	Number of positive cases	Patients included
19 764 tests (=13 179 screened patients)	611	246 (from 08/2013 to 03/2018)

Treatment plan

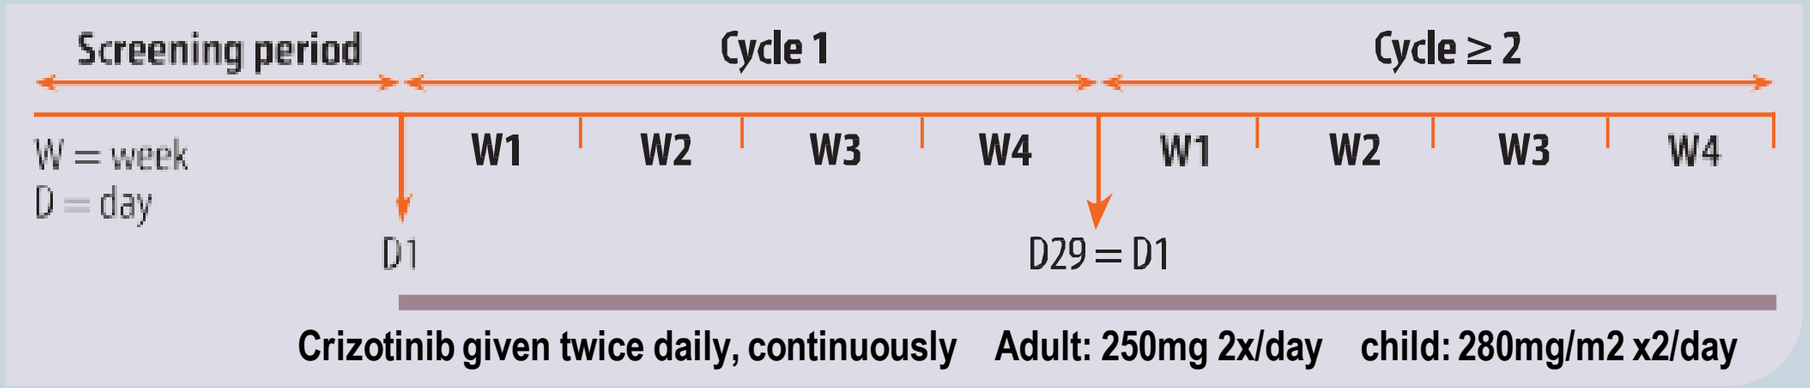

Screening period
Cycle 1
Cycle ≥ 2
W = week
D = day
W1
W2
W3
W4
D1
D29 = D1
Crizotinib given twice daily, continuously Adult: 250mg 2x/day child: 280mg/m2 x2/day

- Treatment pursed until progression, unacceptable toxicity, undercurrent conditions, physician decision or patient refusal
- Safety assessed continuously
- Disease response assessed every 8 weeks with CT and/or PET-CT. All imaging and response evaluation were centrally reviewed and response was evaluated according to Lugano criteria taking into account metabolic response when available or to RECIST 1.1 criteria in patients with only CT-based evaluation.

Statistical design

- Main endpoint: objective response (CR, PR) after 2 cycles. May be changed to the best response for cohorts that are displaying delayed responses
- 3 statistical 2-stage designs are considered for cohorts to anticipate 3 situations in terms of expected response rate and incidence. Accrual stops if 0 response / N1 pts; else additional pts are recruited up to N
- General case: most of the cohorts
- Optimistic design: ALCL and NSCLC with ROS1 translocation
- Rare diseases: IMT, neuroblastoma, glioblastoma, RMS, cohorts identified from the pangenomic programs

Situation	Design parameters				Decision rule			
	P0	P1	Alpha	Beta	N1	k1	N	k
General case	10%	30%	10%	10%	11	0	25	4
Optimistic case	20%	40%	10%	10%	7	0	37	7
Very rare disease	10%	30%	15%	15%	8	0	20	3

- Sample size estimation: 500 pts.
- Centers: declared 198 - open 186 - recruiting 85.
- Population: 246 pts inclued (including 27 children). Median age : 58 years old [1-92].

Inclusions per cohorts

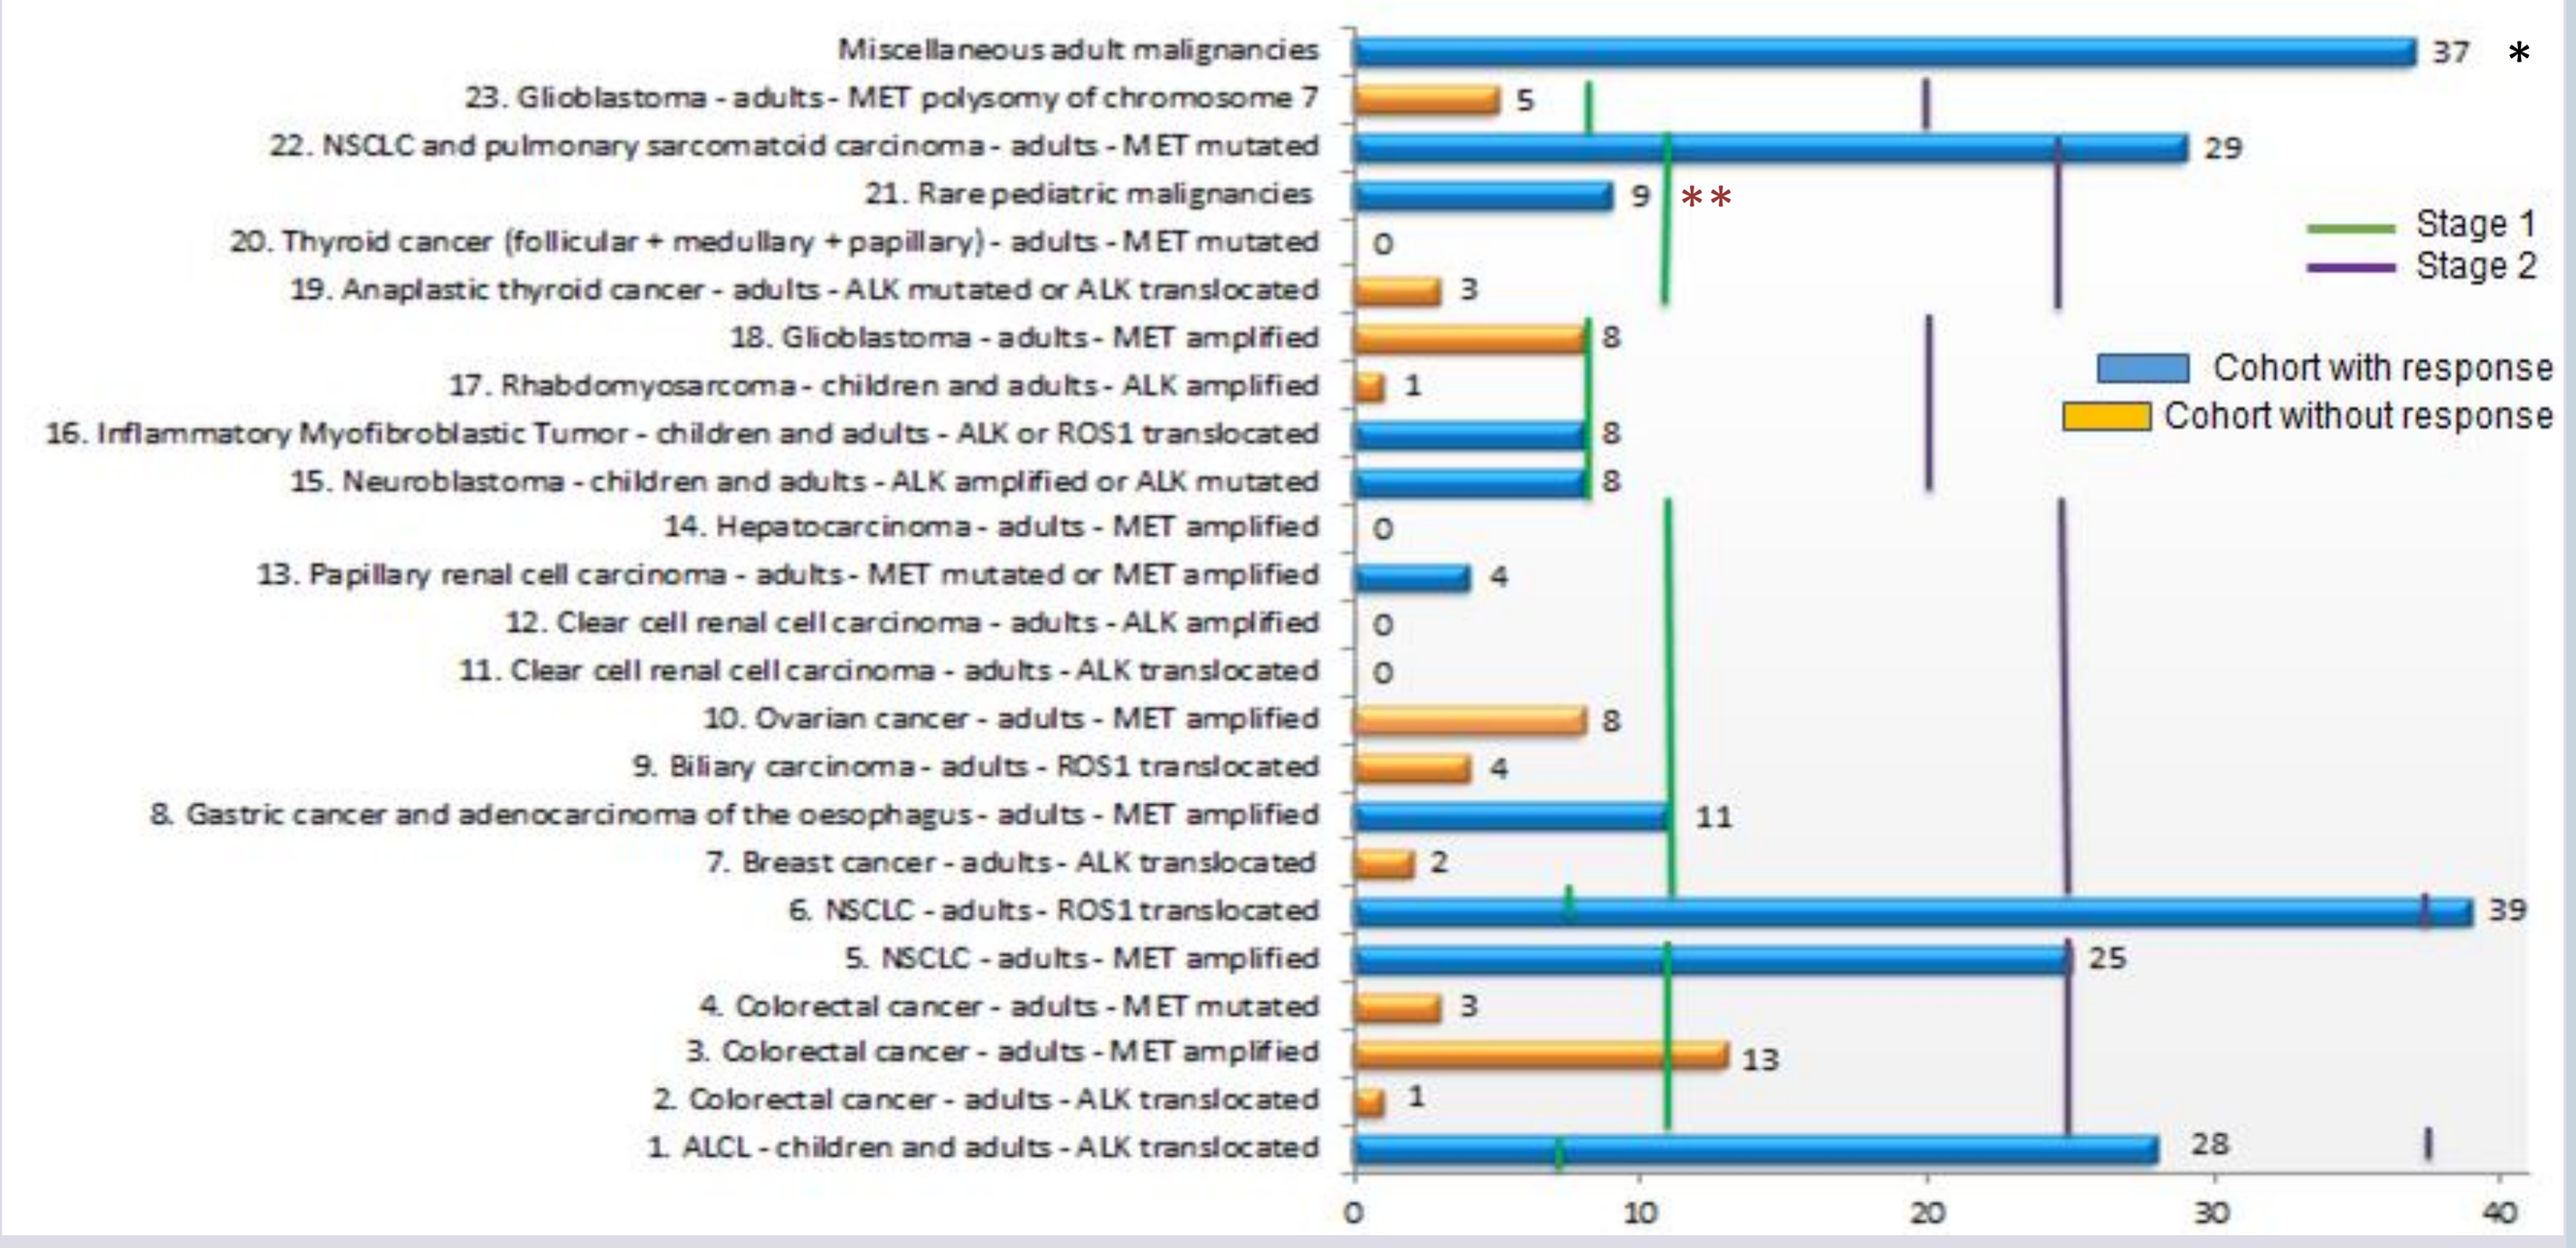

Miscellaneous adult malignancies 37 *
23. Glioblastoma - adults - MET polysomy of chromosome 7 5
22. NSCLC and pulmonary sarcomatoid carcinoma - adults - MET mutated 29
21. Rare pediatric malignancies 9 **
20. Thyroid cancer (follicular + medullary + papillary) - adults - MET mutated 0
19. Anaplastic thyroid cancer - adults - ALK mutated or ALK translocated 3
18. Glioblastoma - adults - MET amplified 8
17. Rhabdomyosarcoma - children and adults - ALK amplified 1
16. Inflammatory Myofibroblastic Tumor - children and adults - ALK or ROS1 translocated 8
15. Neuroblastoma - children and adults - ALK amplified or ALK mutated 8
14. Hepatocarcinoma - adults - MET amplified 0
13. Papillary renal cell carcinoma - adults - MET mutated or MET amplified 4
12. Clear cell renal cell carcinoma - adults - ALK amplified 0
11. Clear cell renal cell carcinoma - adults - ALK translocated 0
10. Ovarian cancer - adults - MET amplified 8
9. Biliary carcinoma - adults - ROS1 translocated 4
8. Gastric cancer and adenocarcinoma of the oesophagus - adults - MET amplified 11
7. Breast cancer - adults - ALK translocated 2
6. NSCLC - adults - ROS1 translocated 39
5. NSCLC - adults - MET amplified 25
4. Colorectal cancer - adults - MET mutated 3
3. Colorectal cancer - adults - MET amplified 13
2. Colorectal cancer - adults - ALK translocated 1
1. ALCL - children and adults - ALK translocated 28
Stage 1
Stage 2
Cohort with response
Cohort without response
0
10
20
30
40

* 3 NSCLC – ALK amplified
1 NSCLC – ALK mutated
1 SCLC – MET mutated + ROS1 mutated
1 urachal adenocarcinoma – MET amplified
4 cholangiocarcinoma – MET amplified
2 cholangiocarcinoma – ALK translocated
1 cholangiocarcinoma – ALK mutated
1 cholangiocarcinoma – MET mutated
1 gallbladder – MET amplified
1 pancreas cancer – ROS1 mutated
1 pancreas cancer – ALK mutated
1 kidney cancer – ROS1 amplified
1 kidney cancer – ALK translocated
1 bladder cancer – MET translocated
1 prostate cancer – ALK translocated
1 colorectal cancer – ALK mutated
2 leiomyosarcoma – ALK translocated
1 small intestine sarcomatoid cancer – ALK translocated
1 larynx carcinoma– MET amplified
1 neuroblastoma – ROS1 mutated
1 B large cell lymphoma – ALK translocated
1 papillary thyroid carcinoma – ALK translocated
1 ethmoid adenocarcinoma – MET amplified
1 breast cancer – ROS1 translocated
1 ovarian cancer – MET mutated
1 ovarian cancer – ALK mutated
1 melanoma – ALK translocated
1 melanoma – ROS1 translocated
1 face epidermoid carcinoma – MET amplified
1 unknown primary carcinoma – ALK translocated.

** 1 mesothelioma – ALK translocated
1 atypical meningioma – ROS1 translocated
1 sarcoma – MET translocated
1 glioma – MET translocated
1 glioma – MET amplified
1 infiltrating glioma – MET amplified
1 liposarcoma – ETV6 NTRK alteration
1 medullary glioma – MET amplified and MET translocated
1 anaplastic pleomorphic xanthoastrocytoma – MET amplified

RESULTS

Biomarkers testing

	ALK Amp	ALK Mut	ALK trans	MET Amp	MET Mut	ROS1 trans
Colorectal	2/788 (0,3%)		2/874 (0,2%)	28/939 (3%)	15/1388 (1%)	
NSCLC				251/4171 (6%)	76/1007 (7,5%)	77/4050 (1,9%)
Breast	5/521 (1%)		2/544 (0,4%)			
Esogastric				35/570 (6%)		
K biliary carcinoma		0/75 (0%)	1/29 (3,4%)	6/107 (5,6%)		4/692 (0,6%)
Ovarian cancer				7/640 (1%)		
Renal carcinoma	0/260 (0%)		0/280 (0%)	3 /83 (3,6%)	9/256 (3,5%)	
Hepatocarcinoma				5/190 (2,6%)		
Neuroblastoma	2/56 (3,6%)	7/65 (11,0%)	1/44 (2,3%)			
IMT			4/12 (33%)			1/4 (25%)
Rhabomyosarcoma	1/44 (2,3%)		2/42 (4,8%)			
Glioblastoma				60/1232 (4,9%)		
Thyroid	0/189 (0%)	1/173 (0,6%)	2/236 (0,8%)		2/185 (1,1%)	
Spitzoid melanoma			0/9 (0%)			0/9 (0%)
	10/1858 (0,5%)	8/313 (2,5%)	14/2070 (0,7%)	395/7932 (5%)	102/2836 (3,6%)	82/4755 (1,7%)

Efficacy

Positive cohorts		Pts analysed	CR/PR at 2 months	ORR% [CI 95%] at 2 months	Best response (CR/PR)	ORR% [CI 95%] best response
ALCL	ALK trans	24	12	54 [29-71]	12	54 [29-71]
NSCLC	MET amp	25	4	16 [4.5-36.1]	8	32 [15-54]
	ROS1 trans	37	17	47.2 [30-64]	25	67.6 [50-82]
	MET mut	28	3	11 [2-28]	10	40 [21-61]
Esogastric	MET amp	9	3	33 [7-70]	5	56 [21-86]
IMT	ALK trans / ROS1 trans	8	3	38 [9-76]	6	75 [35-97]
Neuroblastoma	ALK amp or mut	8	-	-	1	13 [0.3-53]
Kidney	MET amp or mut	4	-	-	1	25 [1-81]
Miscellaneous	pediatrics	9	0	-	3*	
	adults	37	1**	-	1**	

*Sarcoma ETV6NTRK , Mesothelioma ALK trans, Meningioma ROS1 trans
**Kidney ALK trans.

Related toxicities

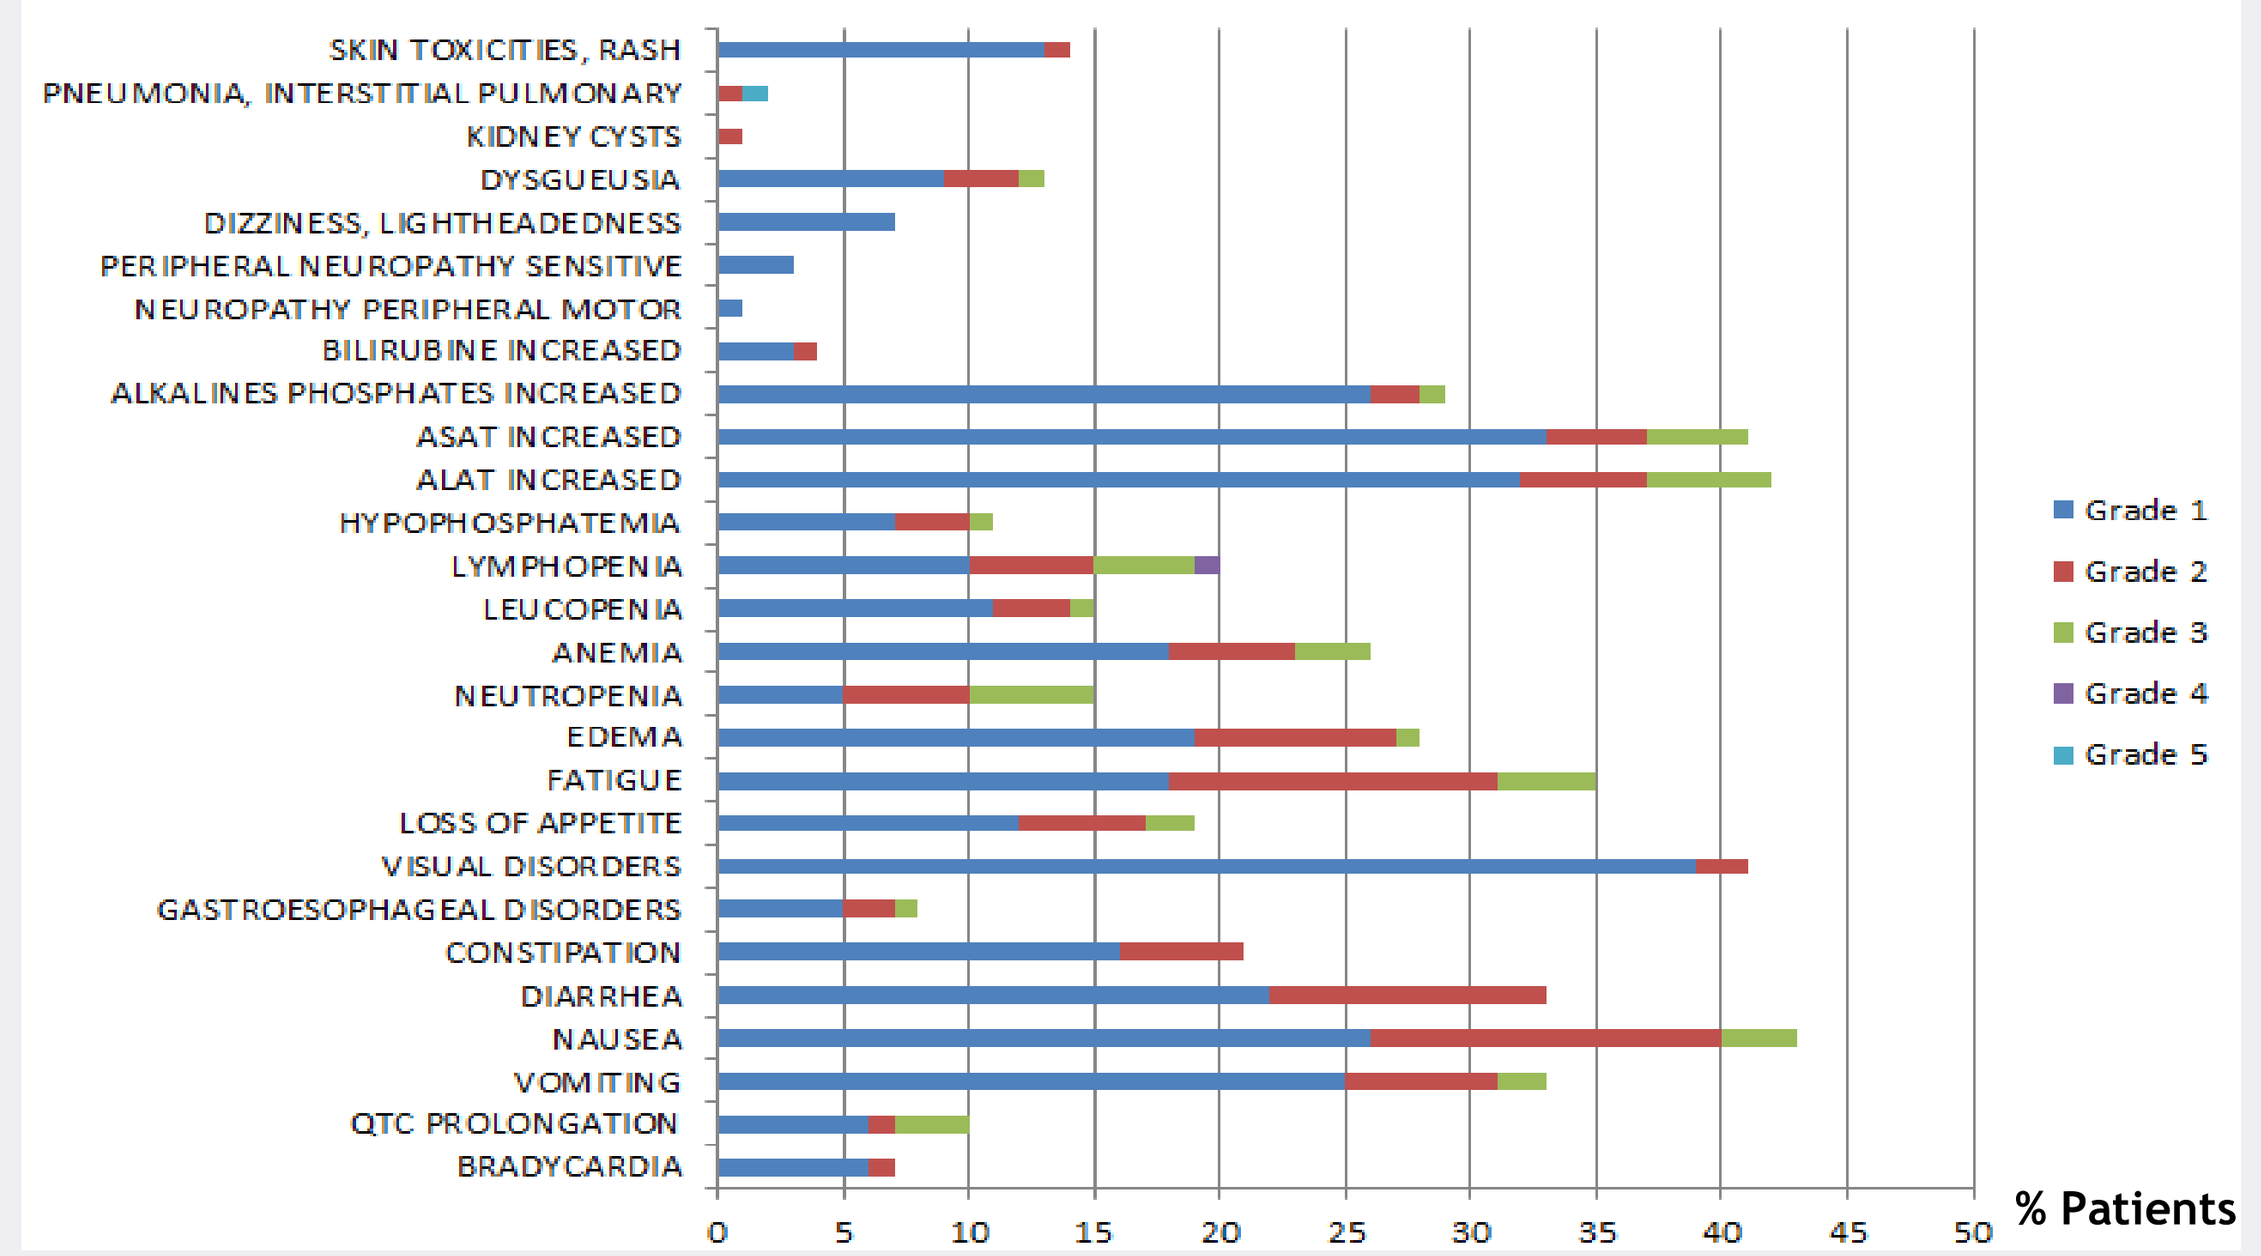

SKIN TOXICITIES, RASH
PNEUMONIA, INTERSTITIAL PULMONARY
KIDNEY CYSTS
DYSGUEUSIA
DIZZINESS, LIGHTHEADEDNESS
PERIPHERAL NEUROPATHY SENSITIVE
NEUROPATHY PERIPHERAL MOTOR
BILIRUBINE INCREASED
ALKALINES PHOSPHATES INCREASED
ASAT INCREASED
ALAT INCREASED
HYPOPHOSPHATEMIA
LYMPHOPENIA
LEUCOPENIA
ANEMIA
NEUTROPENIA
EDEMA
FATIGUE
LOSS OF APPETITE
VISUAL DISORDERS
GASTROESOPHAGEAL DISORDERS
CONSTIPATION
DIARRHEA
NAUSEA
VOMITING
QTC PROLONGATION
BRADYCARDIA
Grade 1
Grade 2
Grade 3
Grade 4
Grade 5
0 5 10 15 20 25 30 35 40 45 50
% Patients

Focus on Non-Small Cell Lung Cancers

MET amplification NSCLC

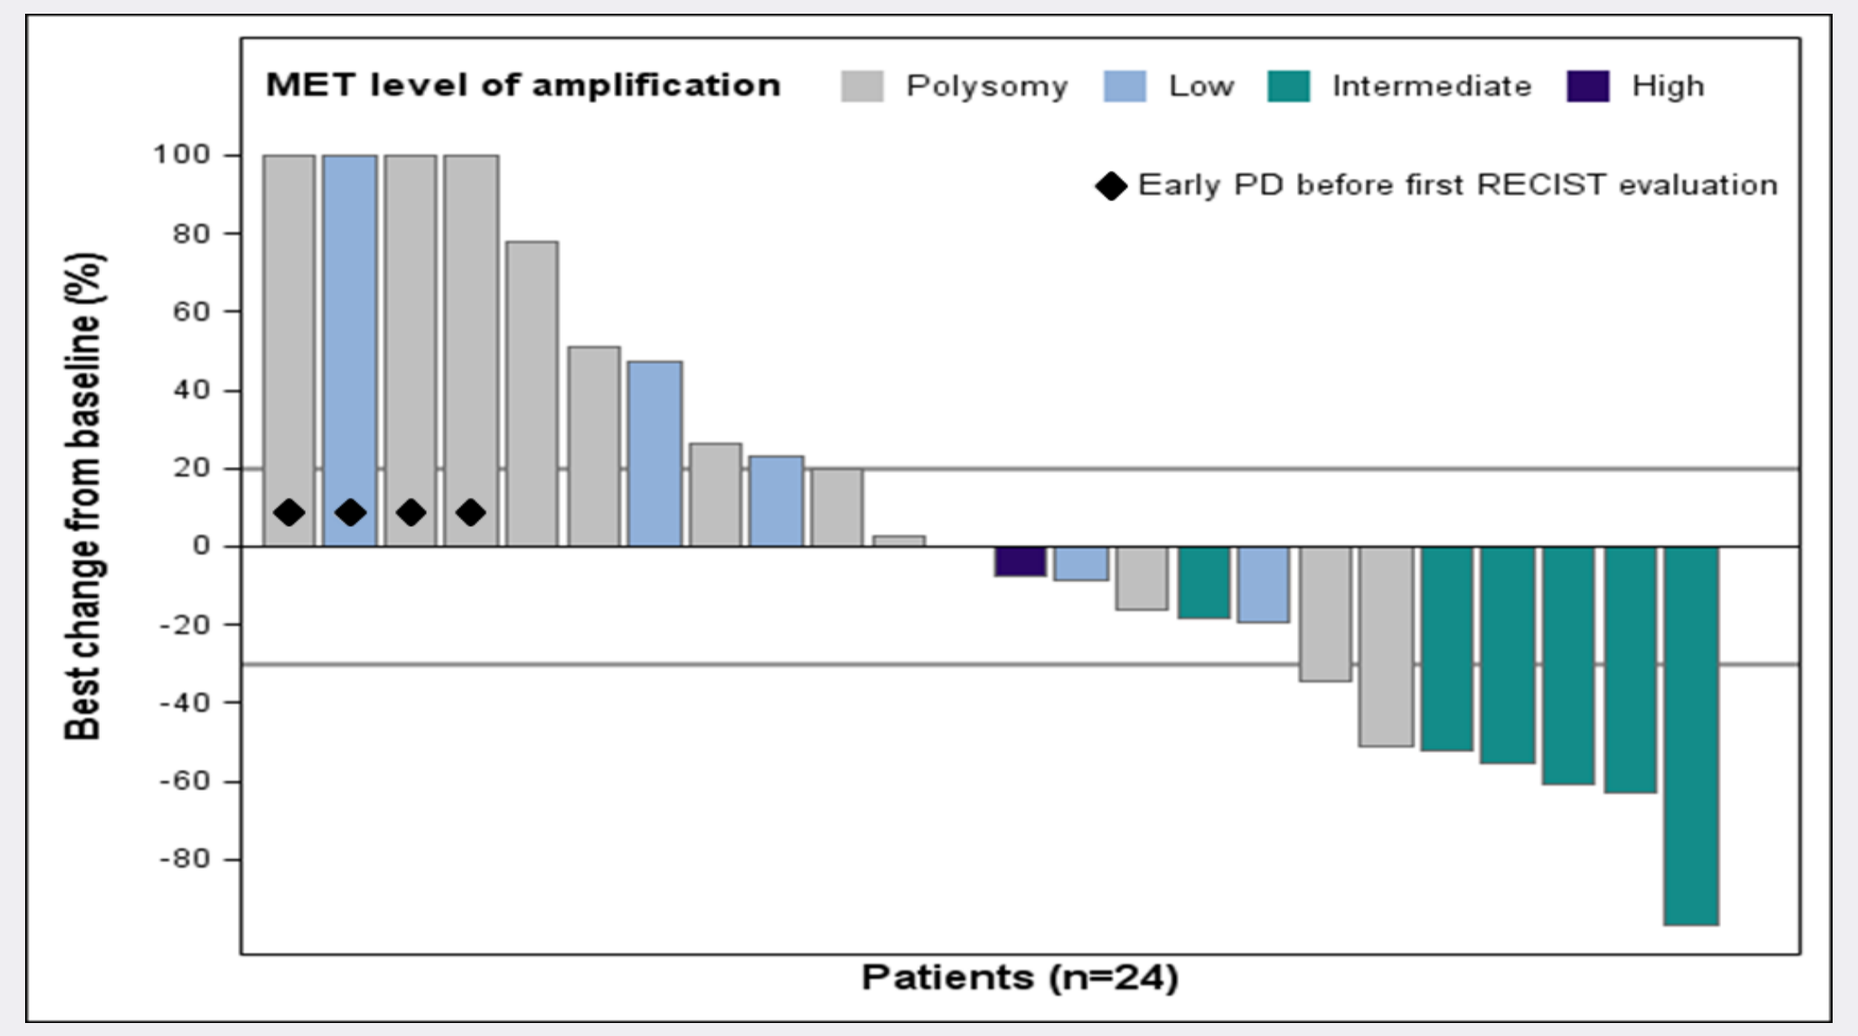

MET level of amplification
Polysomy
Low
Intermediate
High
Early PD before first RECIST evaluation
Best change from baseline (%)
Patients (n=24)

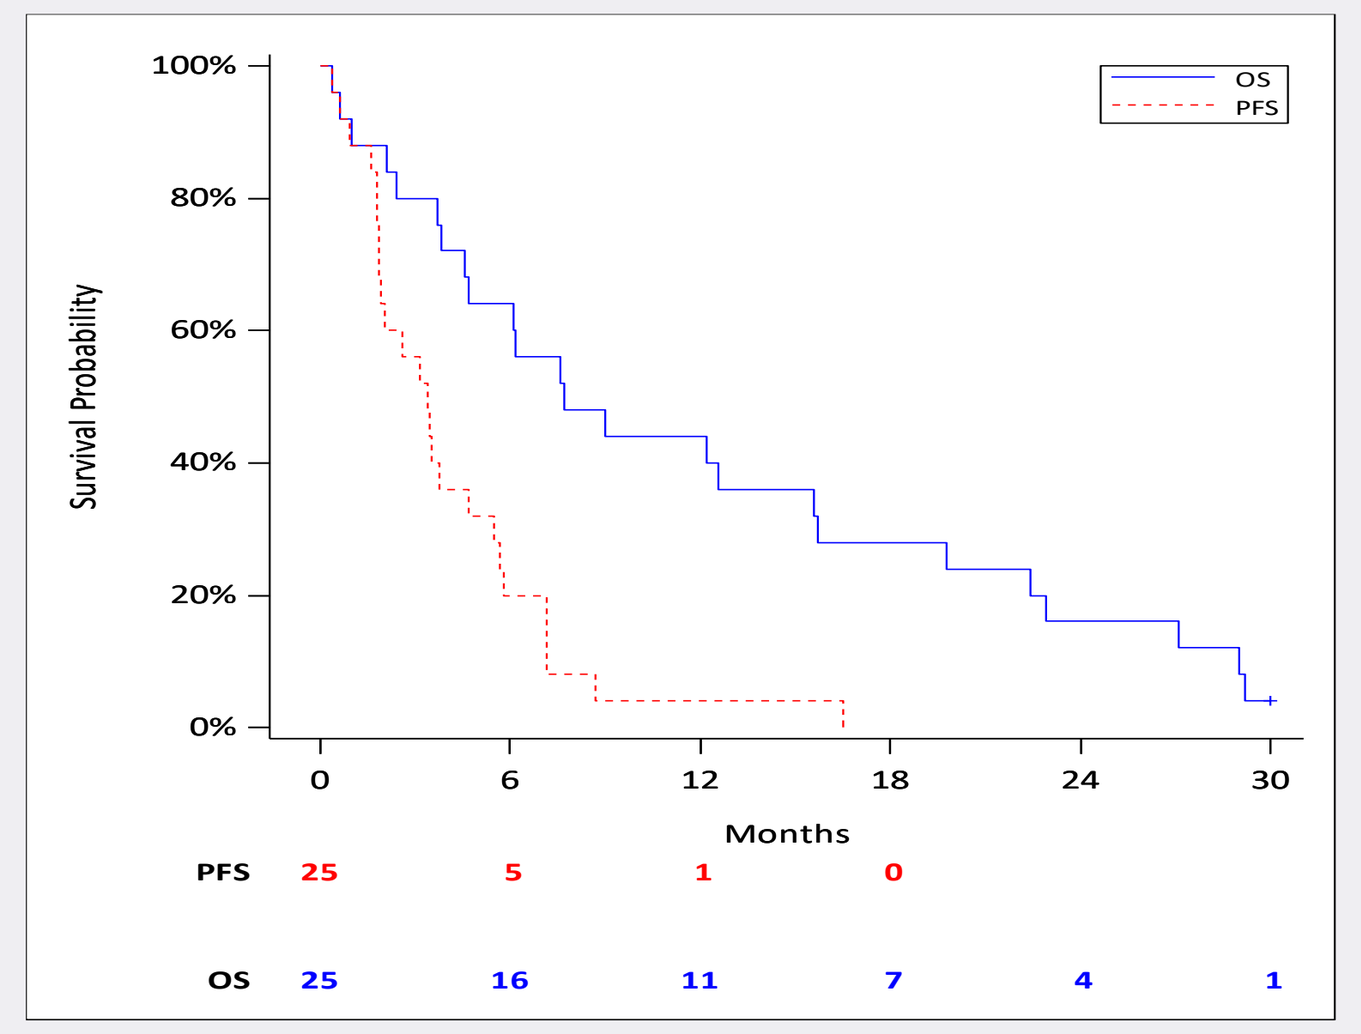

OS
PFS
Survival Probability
Months
PFS 25 5 1 0
OS 25 16 11 7 4 1

MET amplification classification	MET/Centromere ratio
Polysomy	<1.8
Low	≥1.8–≤2.2
Intermediate	>2.2–<5.0
High	≥5.0

Level of *MET* amplification:
- not predictive of response at 2 cycles (CR or PR) p=0.3
- But predictive of best response p=0.04.

Median PFS : 3.4 months 95% CI [1.9; 5.5 months]
Median OS : 7.7 months 95% CI [4.6; 15.7 months]

MET exon 14 mutation NSCLC

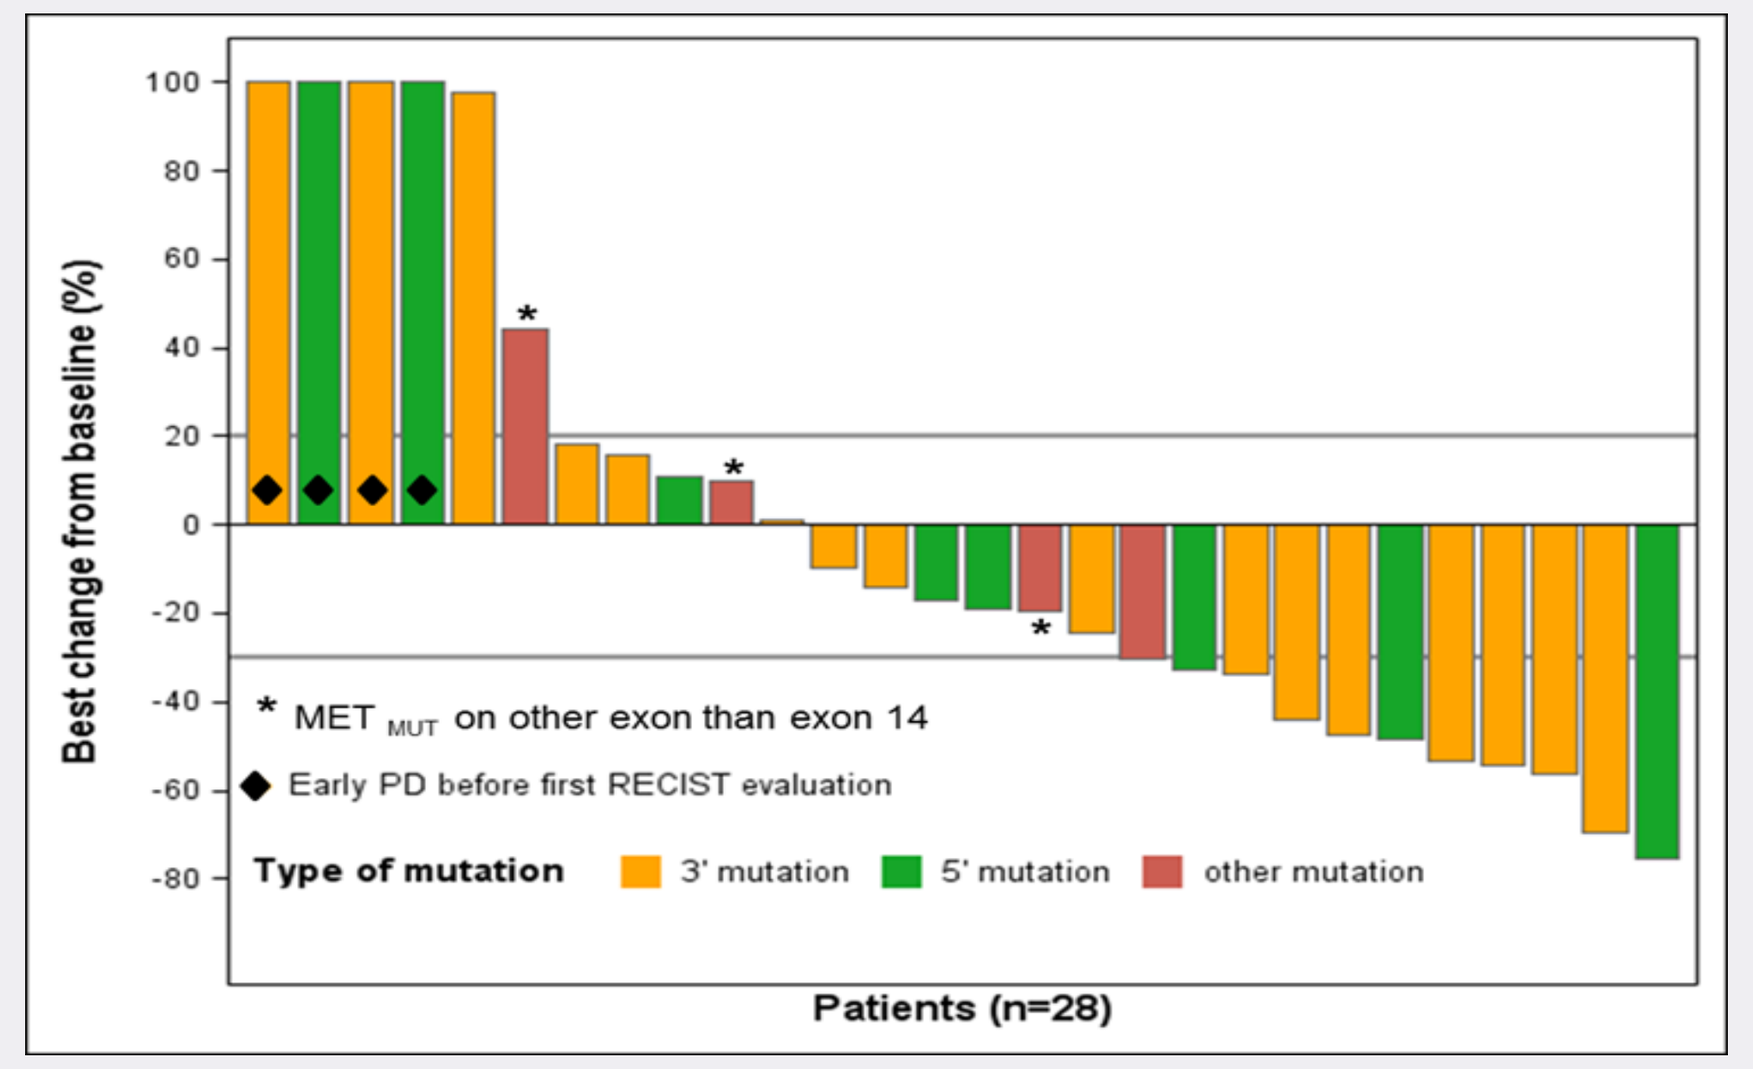

MET MUT on other exon than exon 14
Early PD before first RECIST evaluation
Type of mutation
3' mutation
5' mutation
other mutation
Best change from baseline (%)
Patients (n=28)

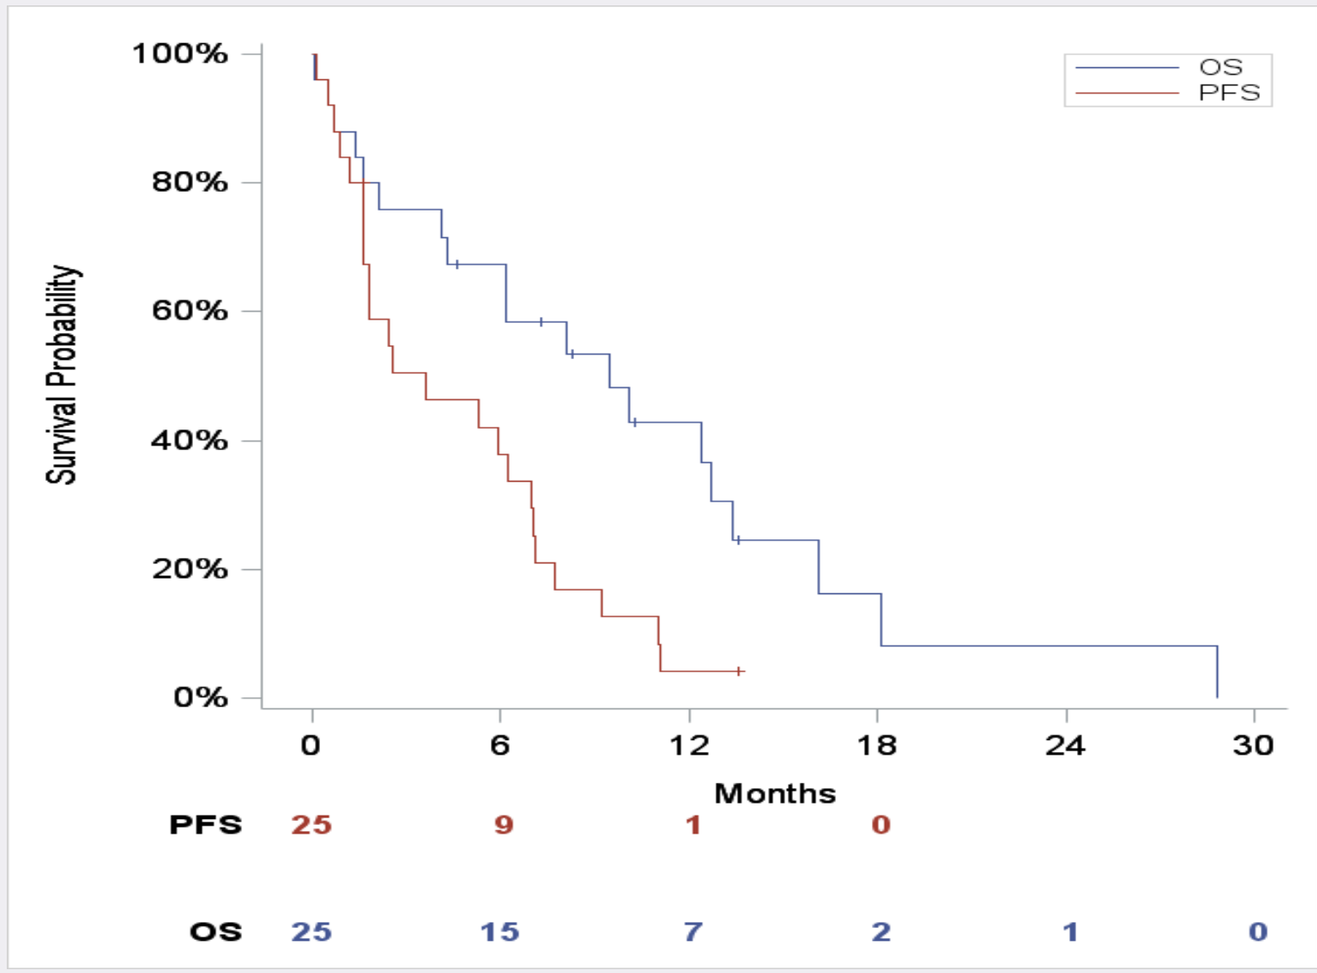

OS
PFS
Survival Probability
Months
PFS 25 9 1 0
OS 25 15 7 2 1 0

Median PFS : 3.6 months with a 95% CI [1.6; 7 months]
Median OS : 9.5 months with a 95% CI [4.1; 13.4months]

ROS1 translocation NSCLC

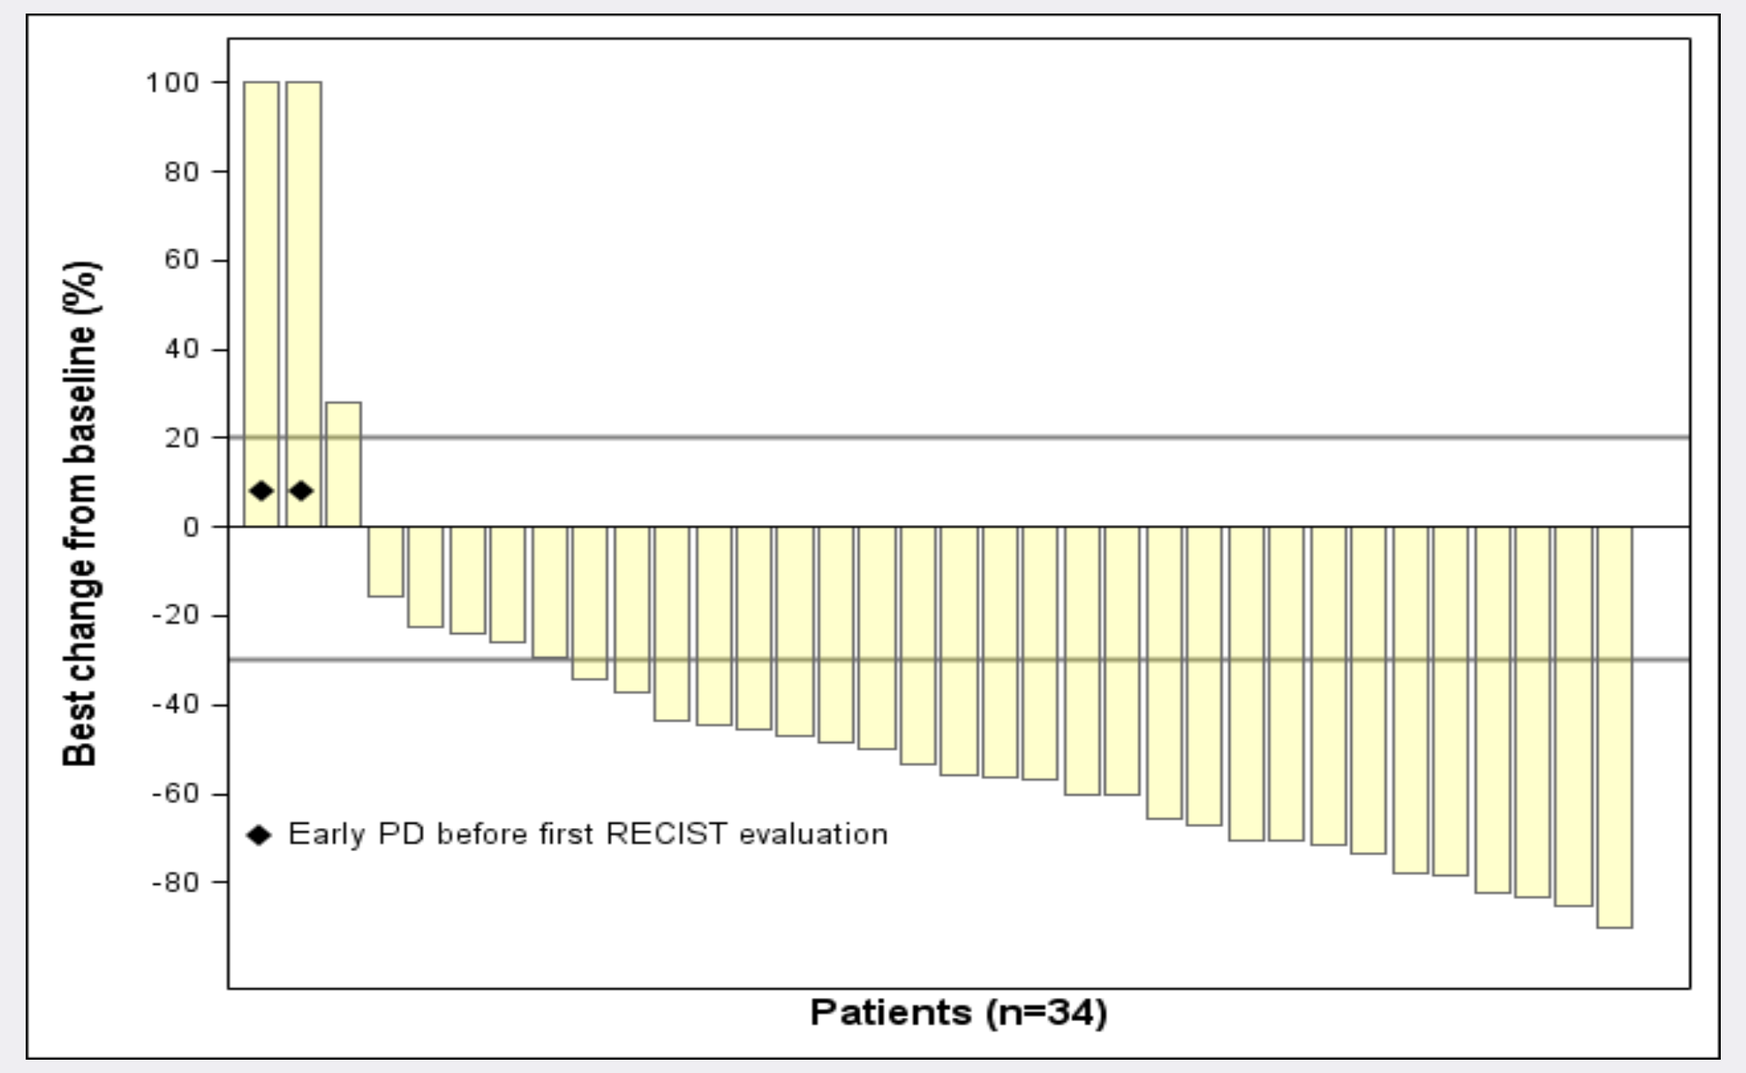

Early PD before first RECIST evaluation
Best change from baseline (%)
Patients (n=34)

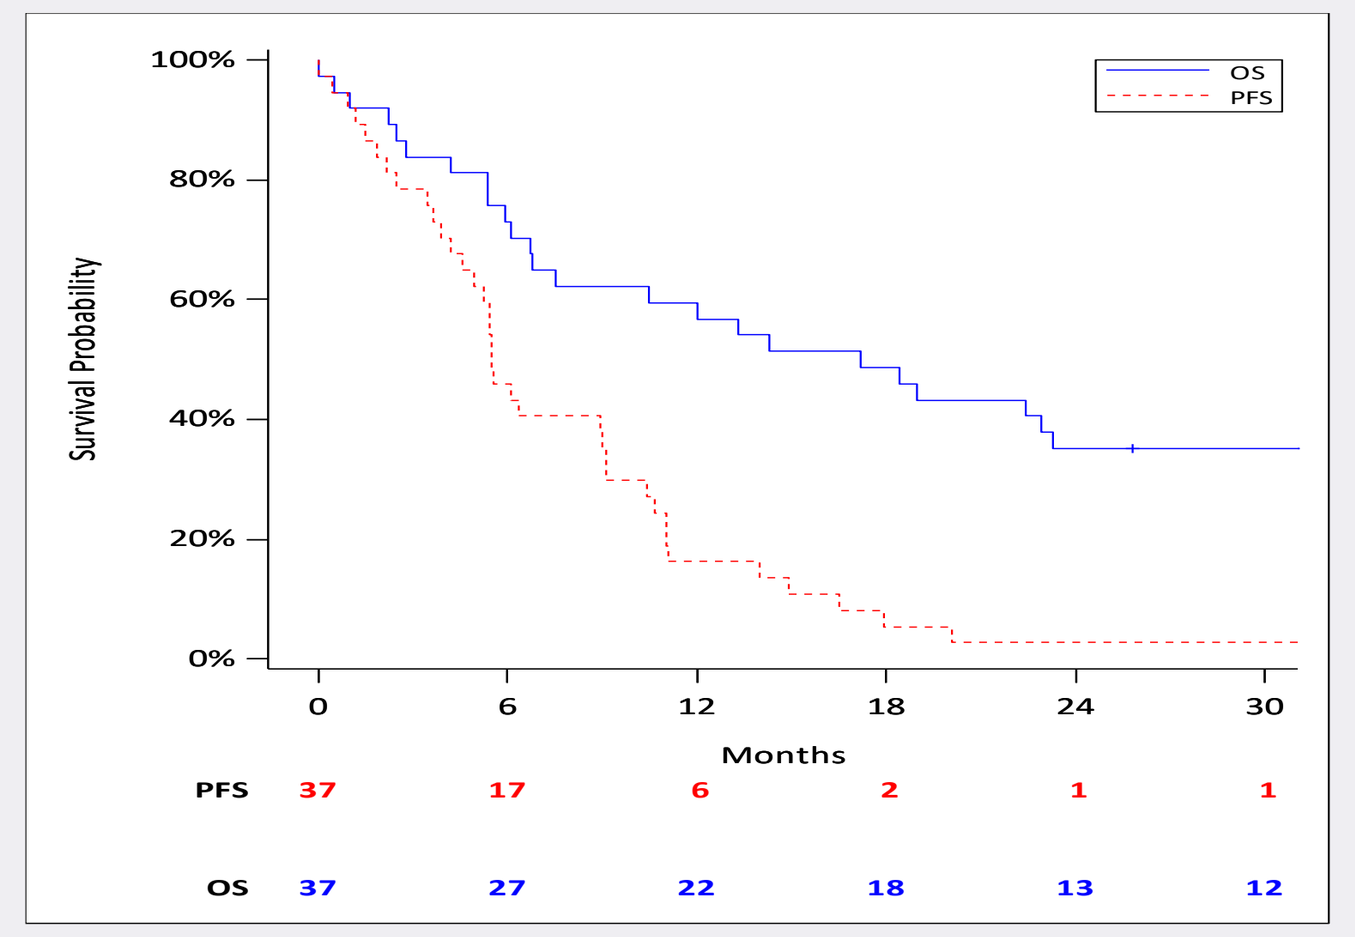

OS
PFS
Survival Probability
Months
PFS 37 17 6 2 1 1
OS 37 27 22 18 13 12

Median PFS : 5.5 months with a 95% CI [4.6; 9.1 months]
Median OS : 17.2 months with a 95% CI [6.8; 32.8 months]

Since start study, crizotinib was registered for ROS1+ stage IV NSCLC (08/2016).

Conclusion

Crizotinib showed activity in pediatric and adults malignancies with ALK, ROS1 and MET alterations.

The AcSé program provided a secured national access to both biomarker testing and treatment for children and adolescents with life threatening malignancies.

Acknowledgements

Patients & families
AcSé crizotinib investigators teams
Study support
- financial support from INCa, the ARC Foundation, the Unicancer's partner for Personalized Medicine research
- institutional support of Pfizer

Collaborative groups
- French thoracic oncology Intergroup (IFCT)
- Francophone Neuro-Oncologist Association (ANOCEF)
- French Cooperative Gynecological Cancer Research Group (ARCAGY-GINECO)
- French Society of Pediatric Oncology (SFCE)
- French Sarcoma Group - Bone Tumor Group (GSF-GETO)
- French Genito-Unirary Cancer Cooperative Groups (GETUG / AFU)
- French Digestive Cancers Groups (UCGI / FFCD / GERCOR)
- French Breast Cancer Intergroup - UNICANCER (UCBG)
- French Thyroid Cancer Network (TUTHYREF)
- Lymphoma Study Association (LYSA)

Correspondence to: Gilles.Vassal@gustaveroussy.fr